Efficacy and Safety of Plecanatide in Patients With Irritable Bowel Syndrome With Constipation: Pooled Analysis of 2 Randomized Clinical Trials

Howard Franklin, MD[1]; Jennifer Christie, MD[2]

[1]Salix Pharmaceuticals, Inc., Bridgewater, NJ, USA; [2]Emory School of Medicine, Atlanta, GA, USA

Scan here to obtain a PDF of this poster.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

4-063

Presented at the ASHP Midyear 2019 Clinical Meeting & Exhibition

December 8–12, 2019

Las Vegas, NV

INTRODUCTION

- Irritable bowel syndrome with constipation (IBS-C) is a chronic condition affecting approximately 5% of the United States population (~16 million people),[1] though prevalence may be underestimated as many people exhibit IBS-C symptoms without a formal diagnosis.[1,2]
- IBS-C is characterized by recurrent abdominal pain related to defecation and/or associated with reduced stool frequency and lumpy/hard stools.[2]
- IBS-C significantly impacts patients' quality of life, work productivity, personal activity, and healthcare expense burden,[1,3-6] with many treated patients reporting low levels of treatment satisfaction with current IBS-C therapies.[3,4,6]
- Plecanatide is structurally identical to human uroguanylin, with the exception of a single amino acid substitution (glutamic acid for aspartic acid in the 3rd position), and both contain 2 disulfide bonds and 2 charged amino acids within the pH-sensitive region. These features are important for the peptide conformation required for binding to the guanylate cyclase-C (GC-C) receptor, and the amino acid substitution in plecanatide enhances the affinity of plecanatide to the GC-C receptor, demonstrating 8 times greater binding to GC-C vs uroguanylin in preclinical models. Based on preclinical studies, plecanatide appears to act in the intestines coinciding with physiological areas of fluid secretion and pain sensation.[7,8]
- Plecanatide is approved in the United States for the treatment of adults with chronic idiopathic constipation and adults with IBS-C.[9]

OBJECTIVE

- To evaluate the efficacy and safety of plecanatide 3 mg and 6 mg once daily in adult patients with IBS-C, through a pooled analysis of two phase 3 clinical trials[10]

METHODS

- Both studies were identically designed, phase 3, randomized, double-blind, placebo-controlled trials of adults with IBS-C in the United States.
- Patients were randomized (1:1:1) to placebo, plecanatide 3 mg, or plecanatide 6 mg, stratified by gender, for 12 weeks of treatment, and a follow-up visit 2 weeks after the last dose of study medication.
- Males and females (aged 18–85 yrs; BMI of 18–40 kg/m^2) meeting Rome III criteria for IBS-C were eligible to participate.
- Patients were ineligible to participate if:
 - They were pregnant or lactating
 - They had any pre-existing medical condition that was considered clinically significant enough to potentially interfere with study assessments or the patient's participation in and completion of the study
- Patients who met the Rome III criteria for IBS-C must also have demonstrated the following during the 2-week pretreatment electronic diary assessment:
 - Completed ≥5 of the 7 daily diary entries in both weeks
 - Reported ≤3 complete spontaneous bowel movements (CSBMs) per week or ≤6 spontaneous bowel movements (SBMs) per week
 - Did not report Bristol Stool Form Scale (BSFS) score of 7 for ≥1 day/week or 6 for >1 day/week for either of the 2 weeks
 - Did not report worst abdominal pain intensity score (11-point numeric rating scale) of 0 for >2 days/week or an average score of <3 for either of the 2 make weeks

Figure 1. Definitions of Responder Endpoints

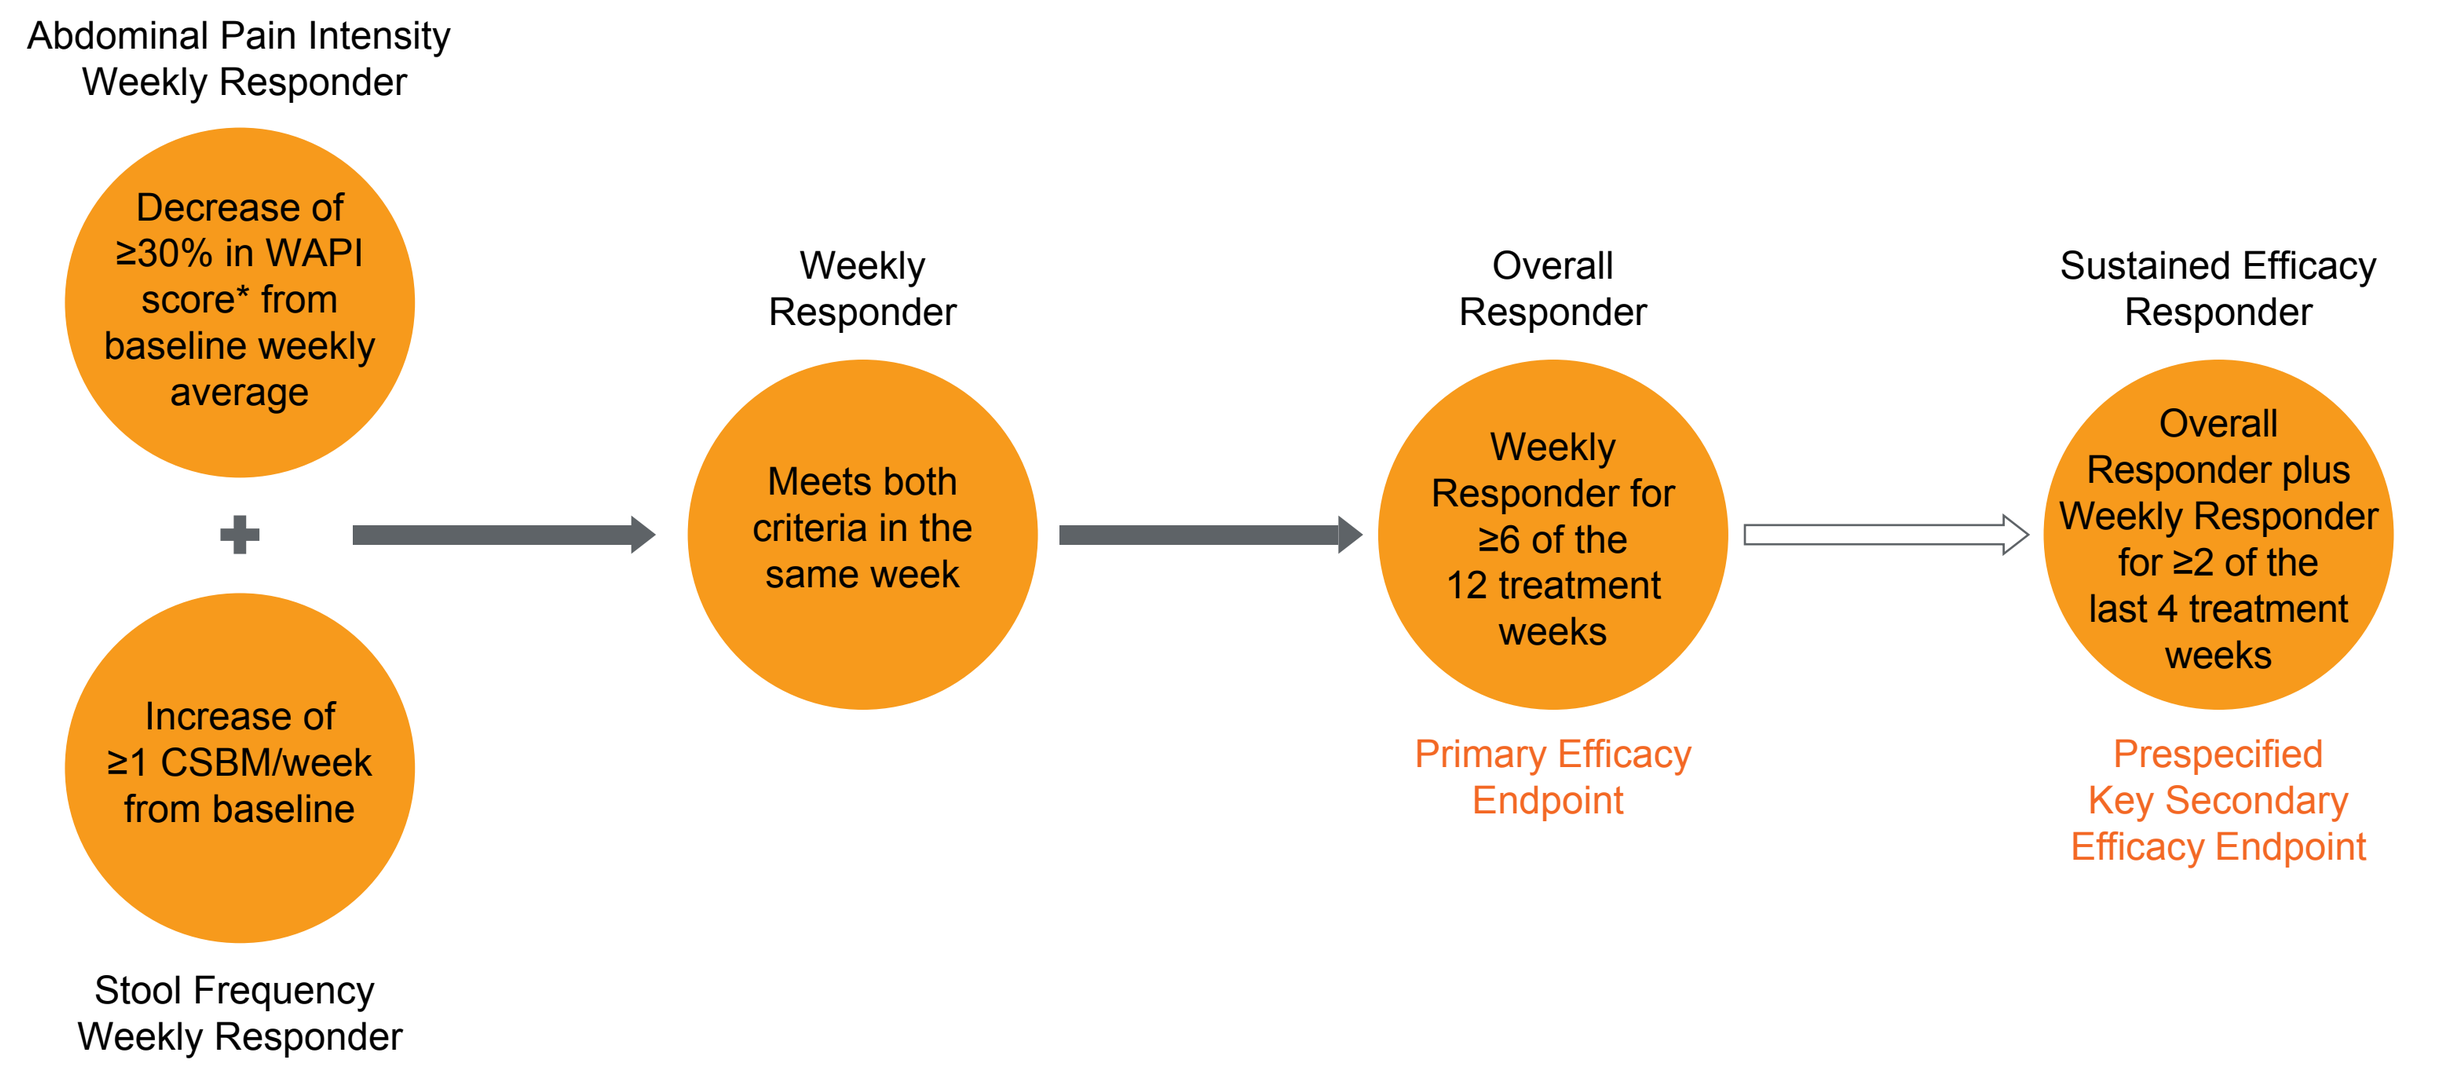
Abdominal Pain Intensity Weekly Responder
Decrease of ≥30% in WAPI score* from baseline weekly average
+
Increase of ≥1 CSBM/week from baseline
Stool Frequency Weekly Responder
Weekly Responder
Meets both criteria in the same week
Overall Responder
Weekly Responder for ≥6 of the 12 treatment weeks
Primary Efficacy Endpoint
Sustained Efficacy Responder
Overall Responder plus Weekly Responder for ≥2 of the last 4 treatment weeks
Prespecified Key Secondary Efficacy Endpoint

*Measured daily. CSBM, complete spontaneous bowel movement; WAPI, worst abdominal pain intensity.

- The primary efficacy endpoint was the percentage of Overall Responders, defined as patients who were Weekly Responders for ≥6 of 12 treatment weeks (Figure 1)
- Key secondary efficacy endpoints included the percentage of Sustained Efficacy Responders (Figure 1), the change from baseline in stool consistency, and the change from baseline in straining severity.
- Other secondary endpoints included:
 - Change from baseline in CSBM frequency rates
 - Change from baseline in the severity of abdominal symptoms
 - Percentage of patients experiencing a CSBM or SBM within 24 hours after the first dose of study medication
 - Mean scores on Patient Global Assessment questionnaires for treatment satisfaction and treatment continuation
- Safety was assessed by the incidence, nature, and severity of adverse events (AEs).
- The primary efficacy analysis and secondary efficacy analyses of the responder endpoints used the Cochran-Mantel-Haenszel test stratified by gender.
- Continuous (change from baseline) efficacy endpoints were analyzed by analysis of covariance, with the model including fixed effects for gender (stratification variable) and treatment, and a covariate for the corresponding baseline value.

RESULTS

Table 1. Demographics and Baseline Characteristics

Patients, n (%)	Placebo (N=733)	Plecanatide 3 mg (N=728)	Plecanatide 6 mg (N=728)
Age, years, mean (range)	43.9 (18–81)	43.5 (18–83)	43.1 (18–83)
Gender			
Female	74.2%	73.8%	74.0%
Male	25.8%	26.2%	26.0%
Race			
White	73.4%	72.7%	71.2%
Black	22.1%	21.4%	24.6%
Other	4.5%	5.9%	4.2%
BMI, kg/m^2, mean (range)	28.0 (18–40)	28.2 (18–40)	28.0 (17–42)
Disease characteristics, mean (SD)			
CSBMs/week	0.23 (0.5)	0.24 (0.5)	0.27 (0.5)
Stool consistency	2.0 (1.0)	2.0 (0.9)	1.9 (0.9)
Straining severity	6.6 (1.9)	6.7 (1.9)	6.7 (1.9)
Abdominal pain	6.3 (1.7)	6.3 (1.7)	6.2 (1.8)

BMI, body mass index; CSBM, complete spontaneous bowel movement; SD, standard deviation.

- 2189 patients were included in the intention-to-treat population, with similar demographics between treatment groups and across studies (Table 1).
- These studies enrolled a large percentage of men (~26%) compared with other phase 3 IBS-C clinical trials and a large number of patients with moderate scores (>6) in straining and abdominal pain severity.

Figure 2. Percentage of Patients Who Were Overall Responders

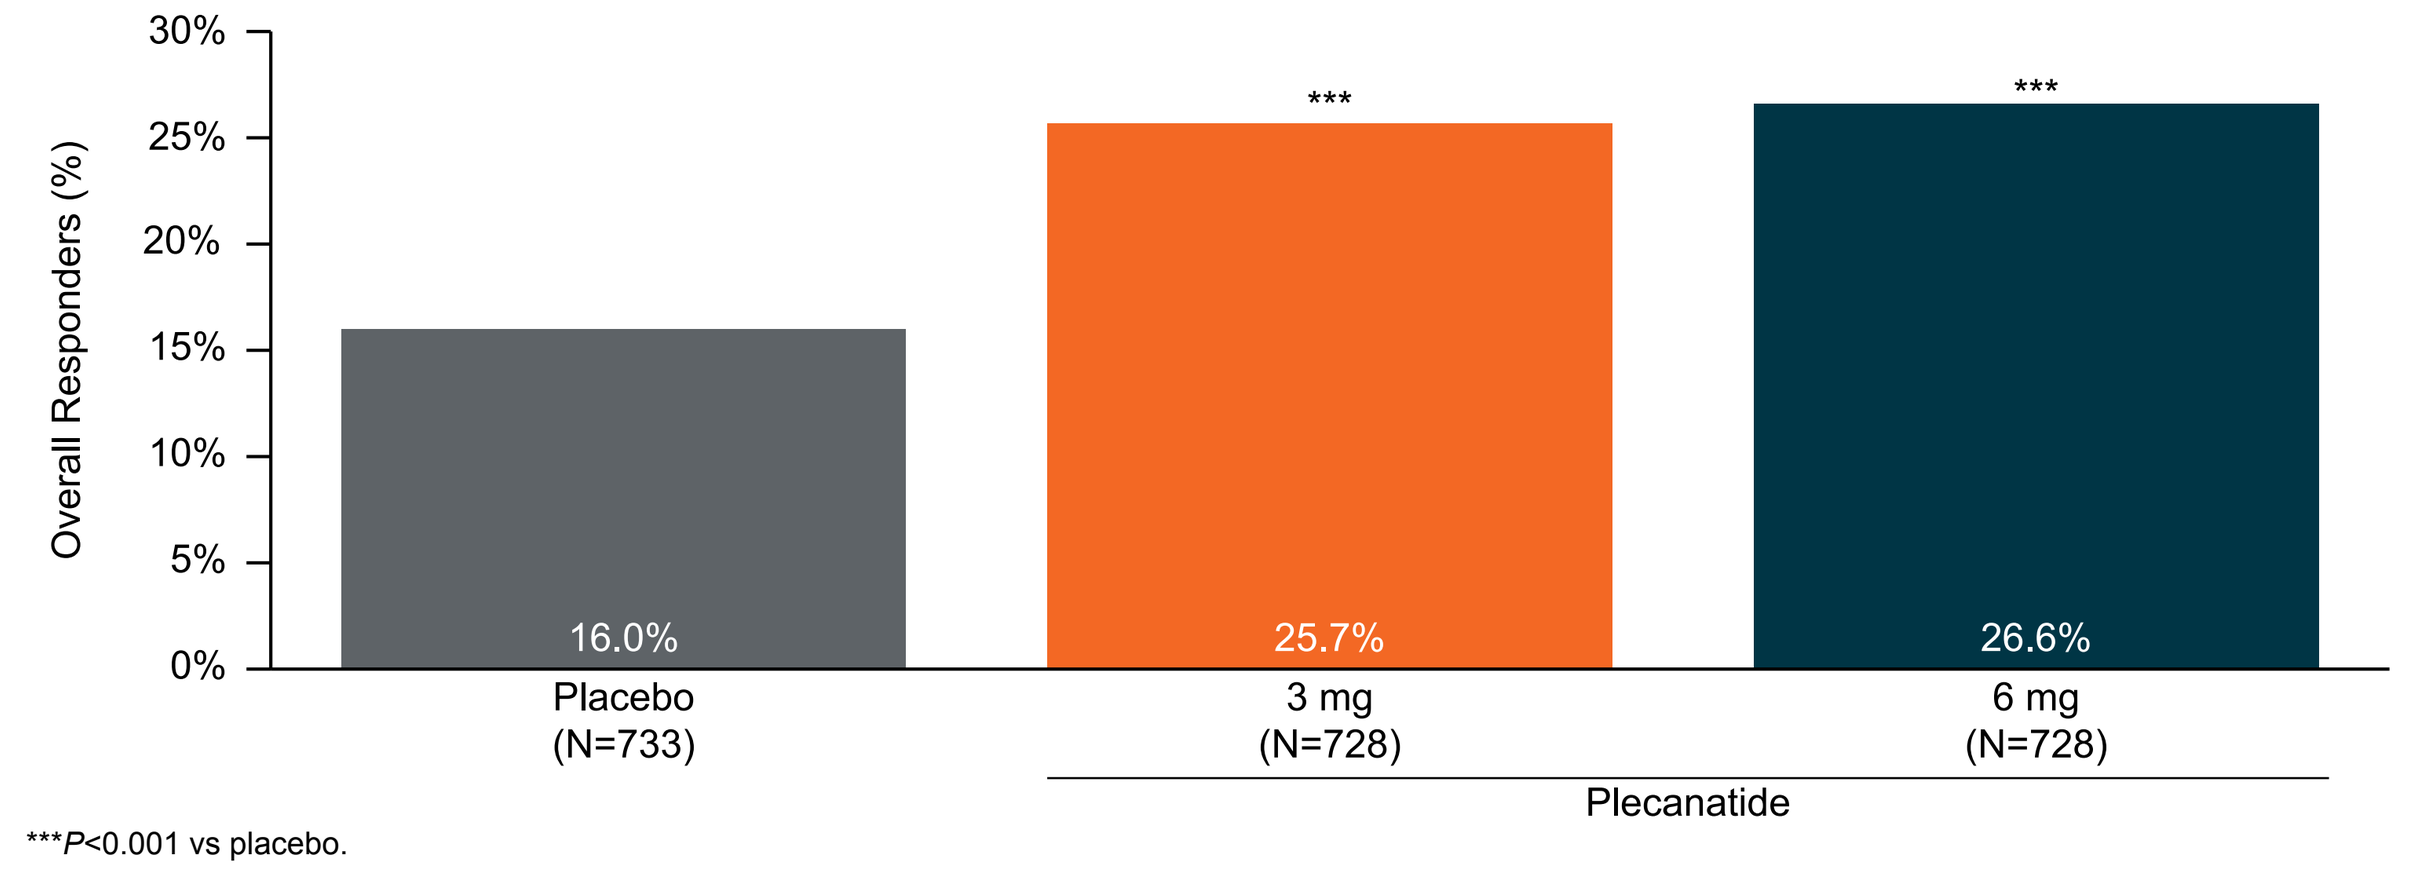
Overall Responders (%)
30%
25%
20%
15%
10%
5%
0%

16.0%
25.7%
26.6%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide

***P<0.001 vs placebo.

- Plecanatide treatment resulted in a significantly greater percentage of Overall Responders compared with placebo (Figure 2).

Figure 3. Percentage of Patients Who Were Weekly Responders by Time Point

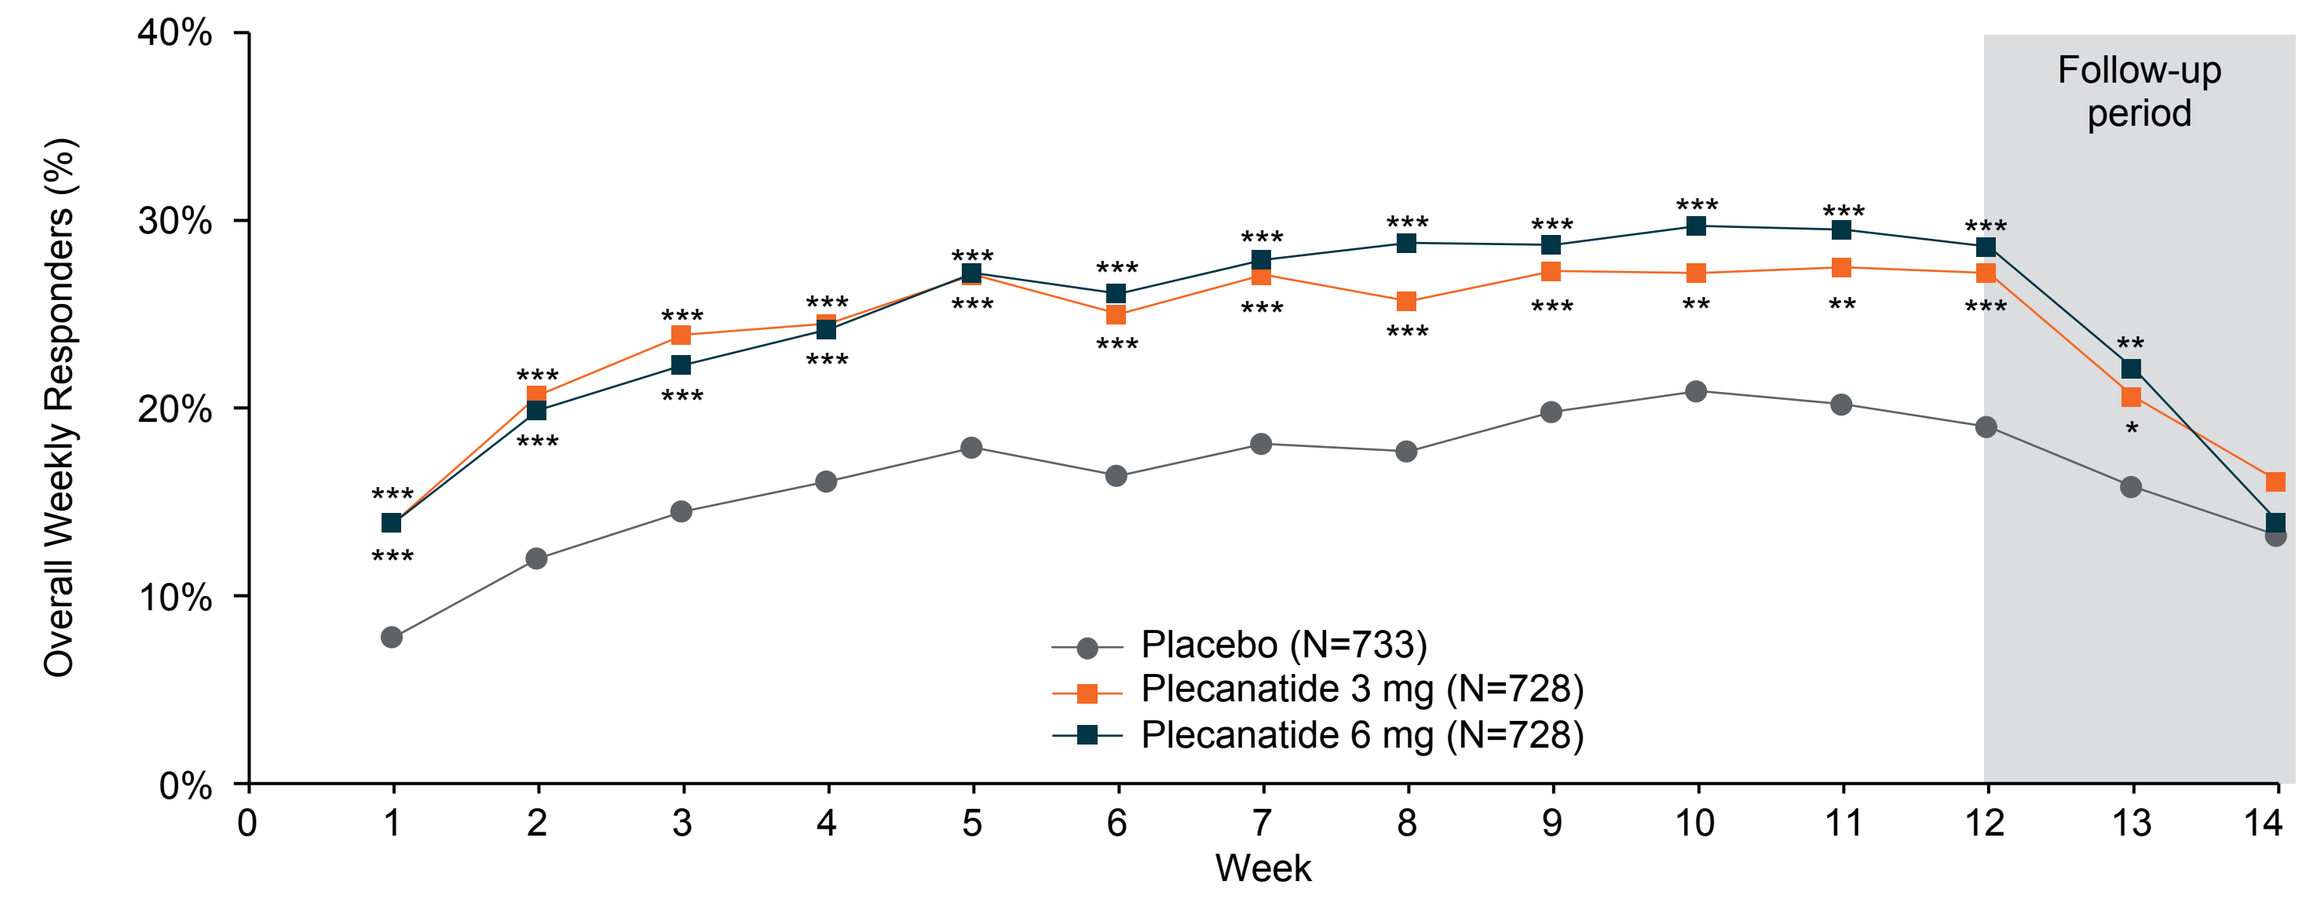
Overall Weekly Responders (%)
40%
30%
20%
10%
0%
Follow-up period
Placebo (N=733)
Plecanatide 3 mg (N=728)
Plecanatide 6 mg (N=728)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14
Week

*P<0.05, **P<0.01, ***P<0.001 vs placebo.

- Significant differences favoring plecanatide over placebo were apparent following the first week of treatment and continued throughout the 12-week treatment period (Figure 3).

Figure 4. Percentage of Patients Who Were Sustained Efficacy Responders

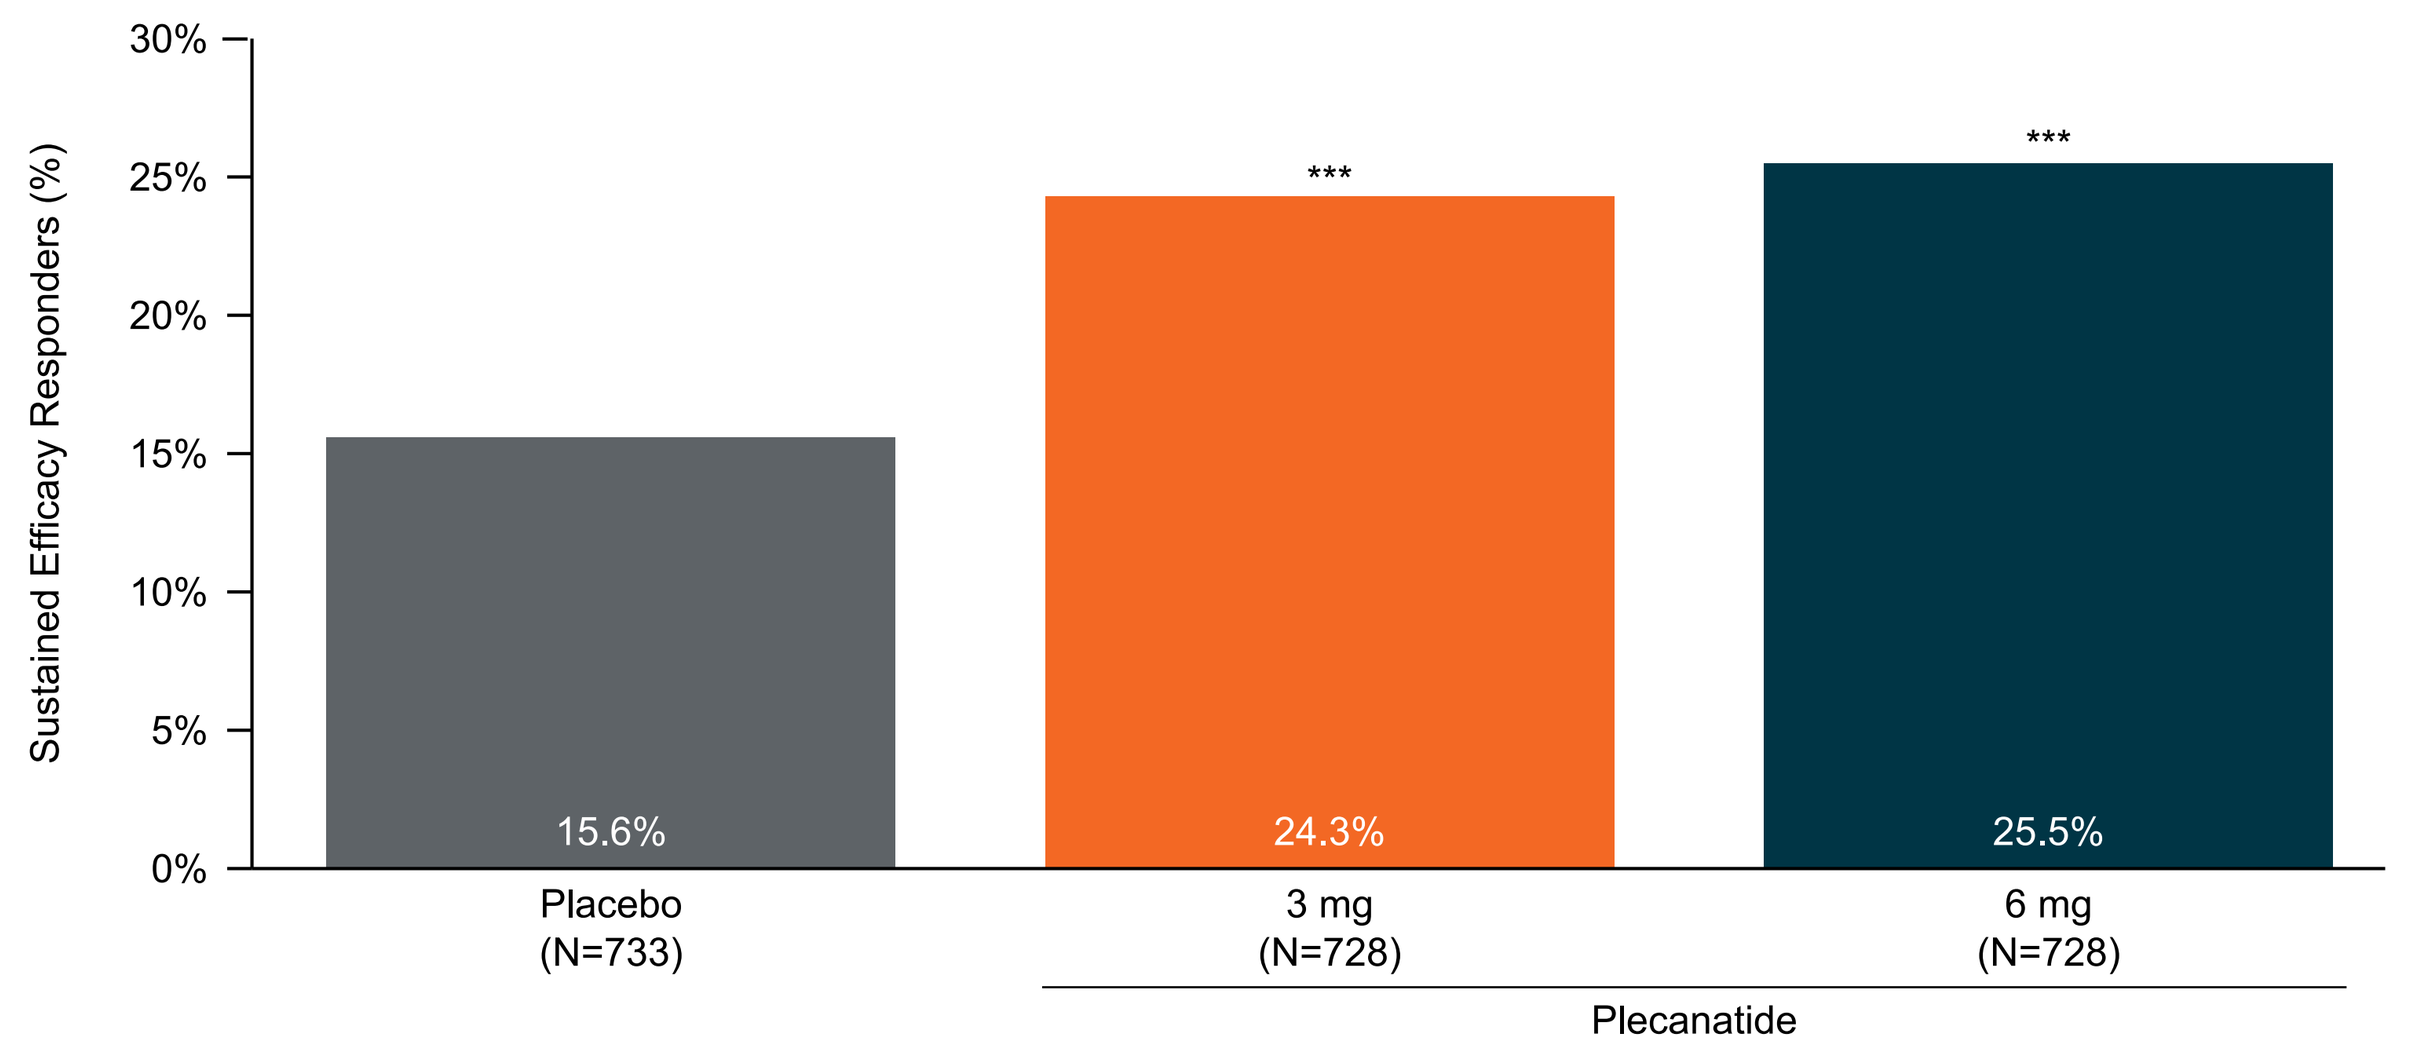
Sustained Efficacy Responders (%)
30%
25%
20%
15%
10%
5%
0%

15.6%
24.3%
25.5%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide

***P<0.001 vs placebo.

- A significantly greater percentage of plecanatide-treated patients were Sustained Efficacy Responders than were placebo-treated patients (Figure 4).

Figure 5. Percentage of Abdominal Pain Intensity Weekly Responders (A) and Stool Frequency Weekly Responders (B) for ≥6 of 12 Treatment Weeks

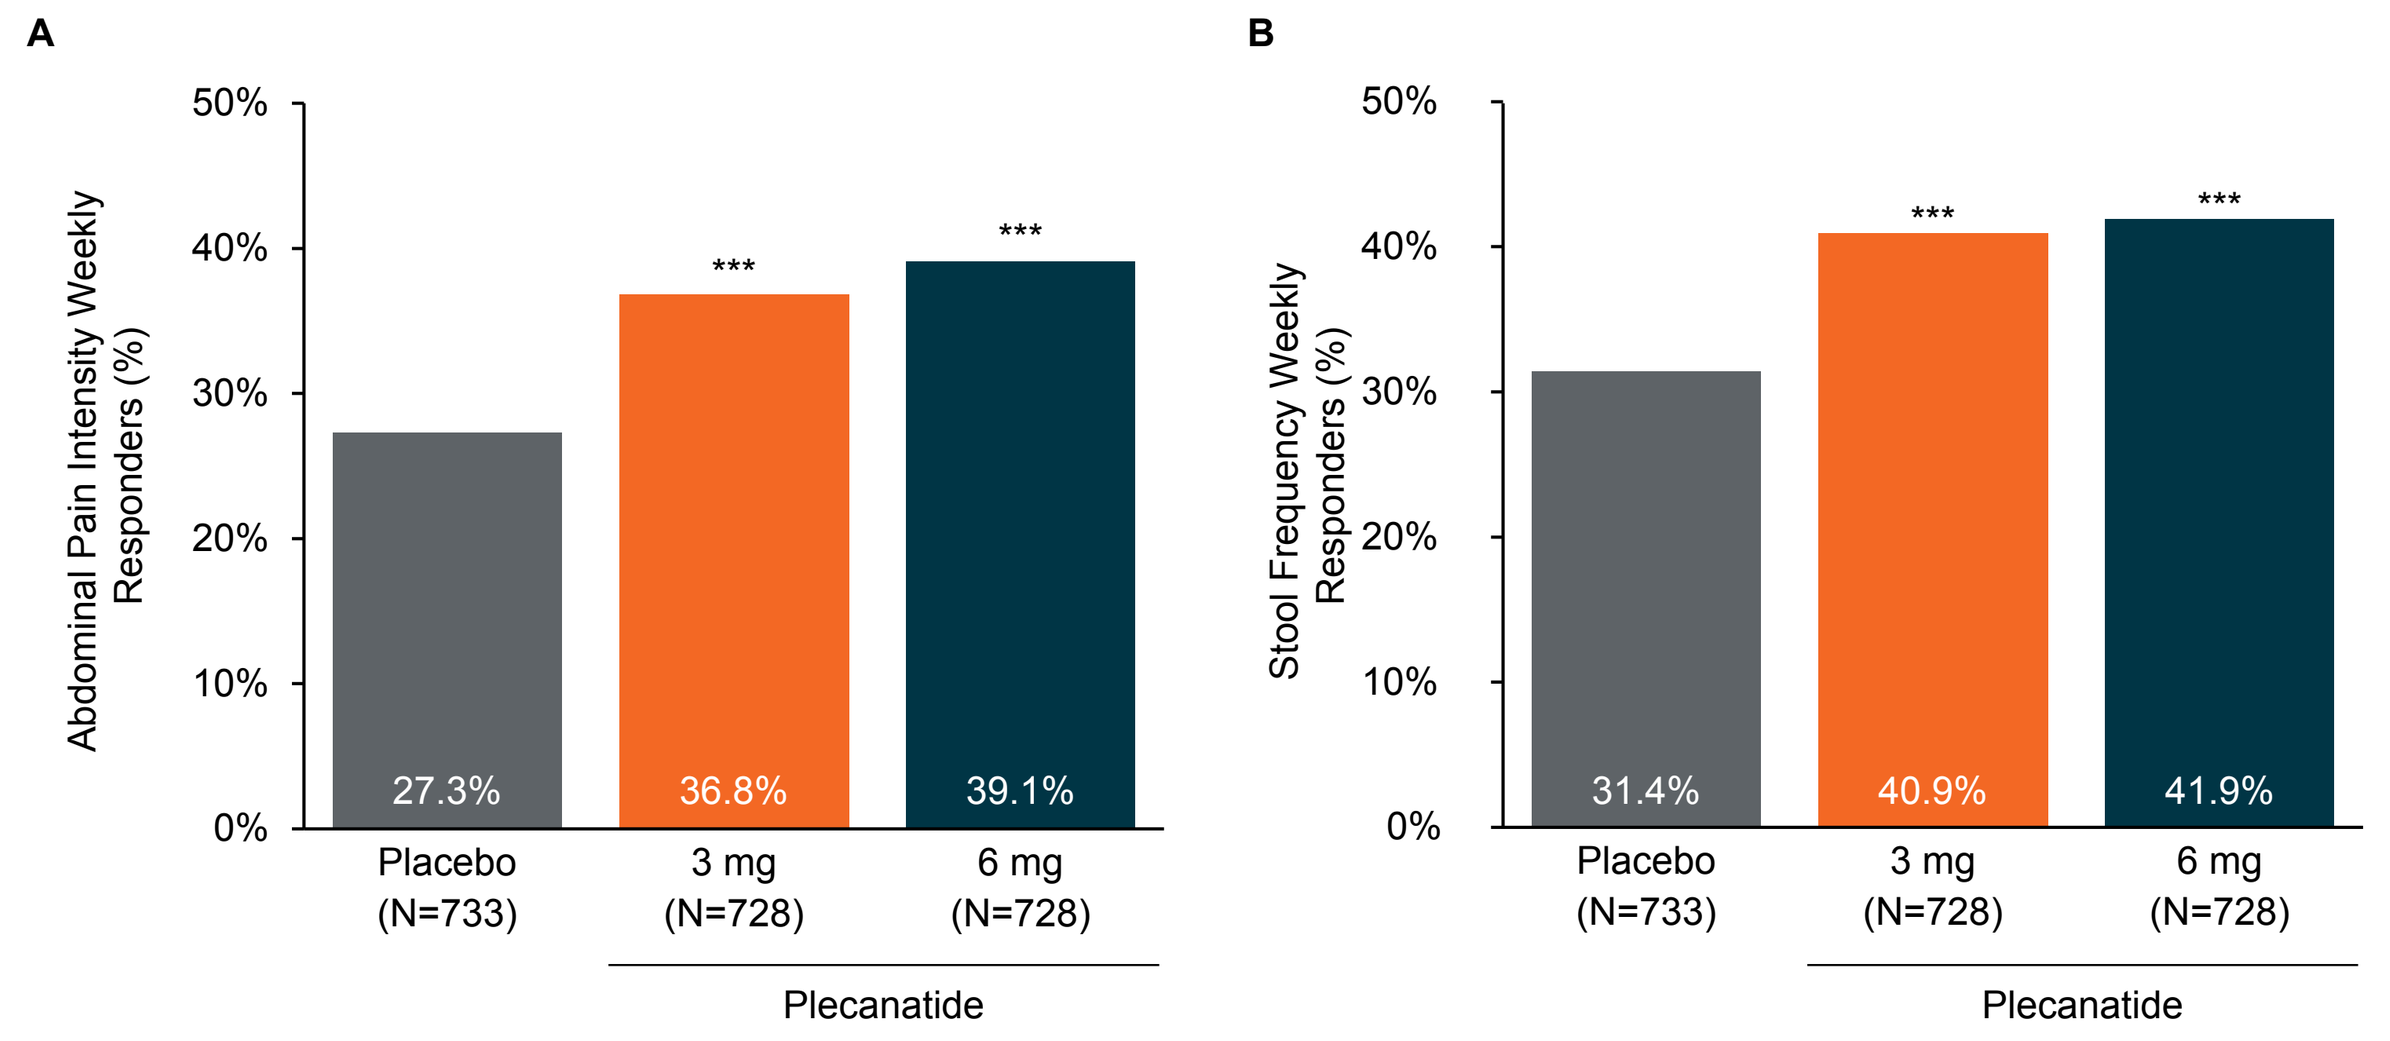
A
Abdominal Pain Intensity Weekly Responders (%)
50%
40%
30%
20%
10%
0%

27.3%
36.8%
39.1%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide
B
Stool Frequency Weekly Responders (%)
50%
40%
30%
20%
10%
0%

31.4%
40.9%
41.9%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide

***P<0.001 vs placebo.

- Plecanatide treatment resulted in a significantly greater percentage of patients who were Abdominal Pain Intensity Weekly Responders (Figure 5A) and Stool Frequency Weekly Responders (Figure 5B) for ≥6 of the 12 weeks compared to placebo.

Figure 6. Percentage of Patients Experiencing a CSBM (A) or SBM (B) Within First 24 Hours

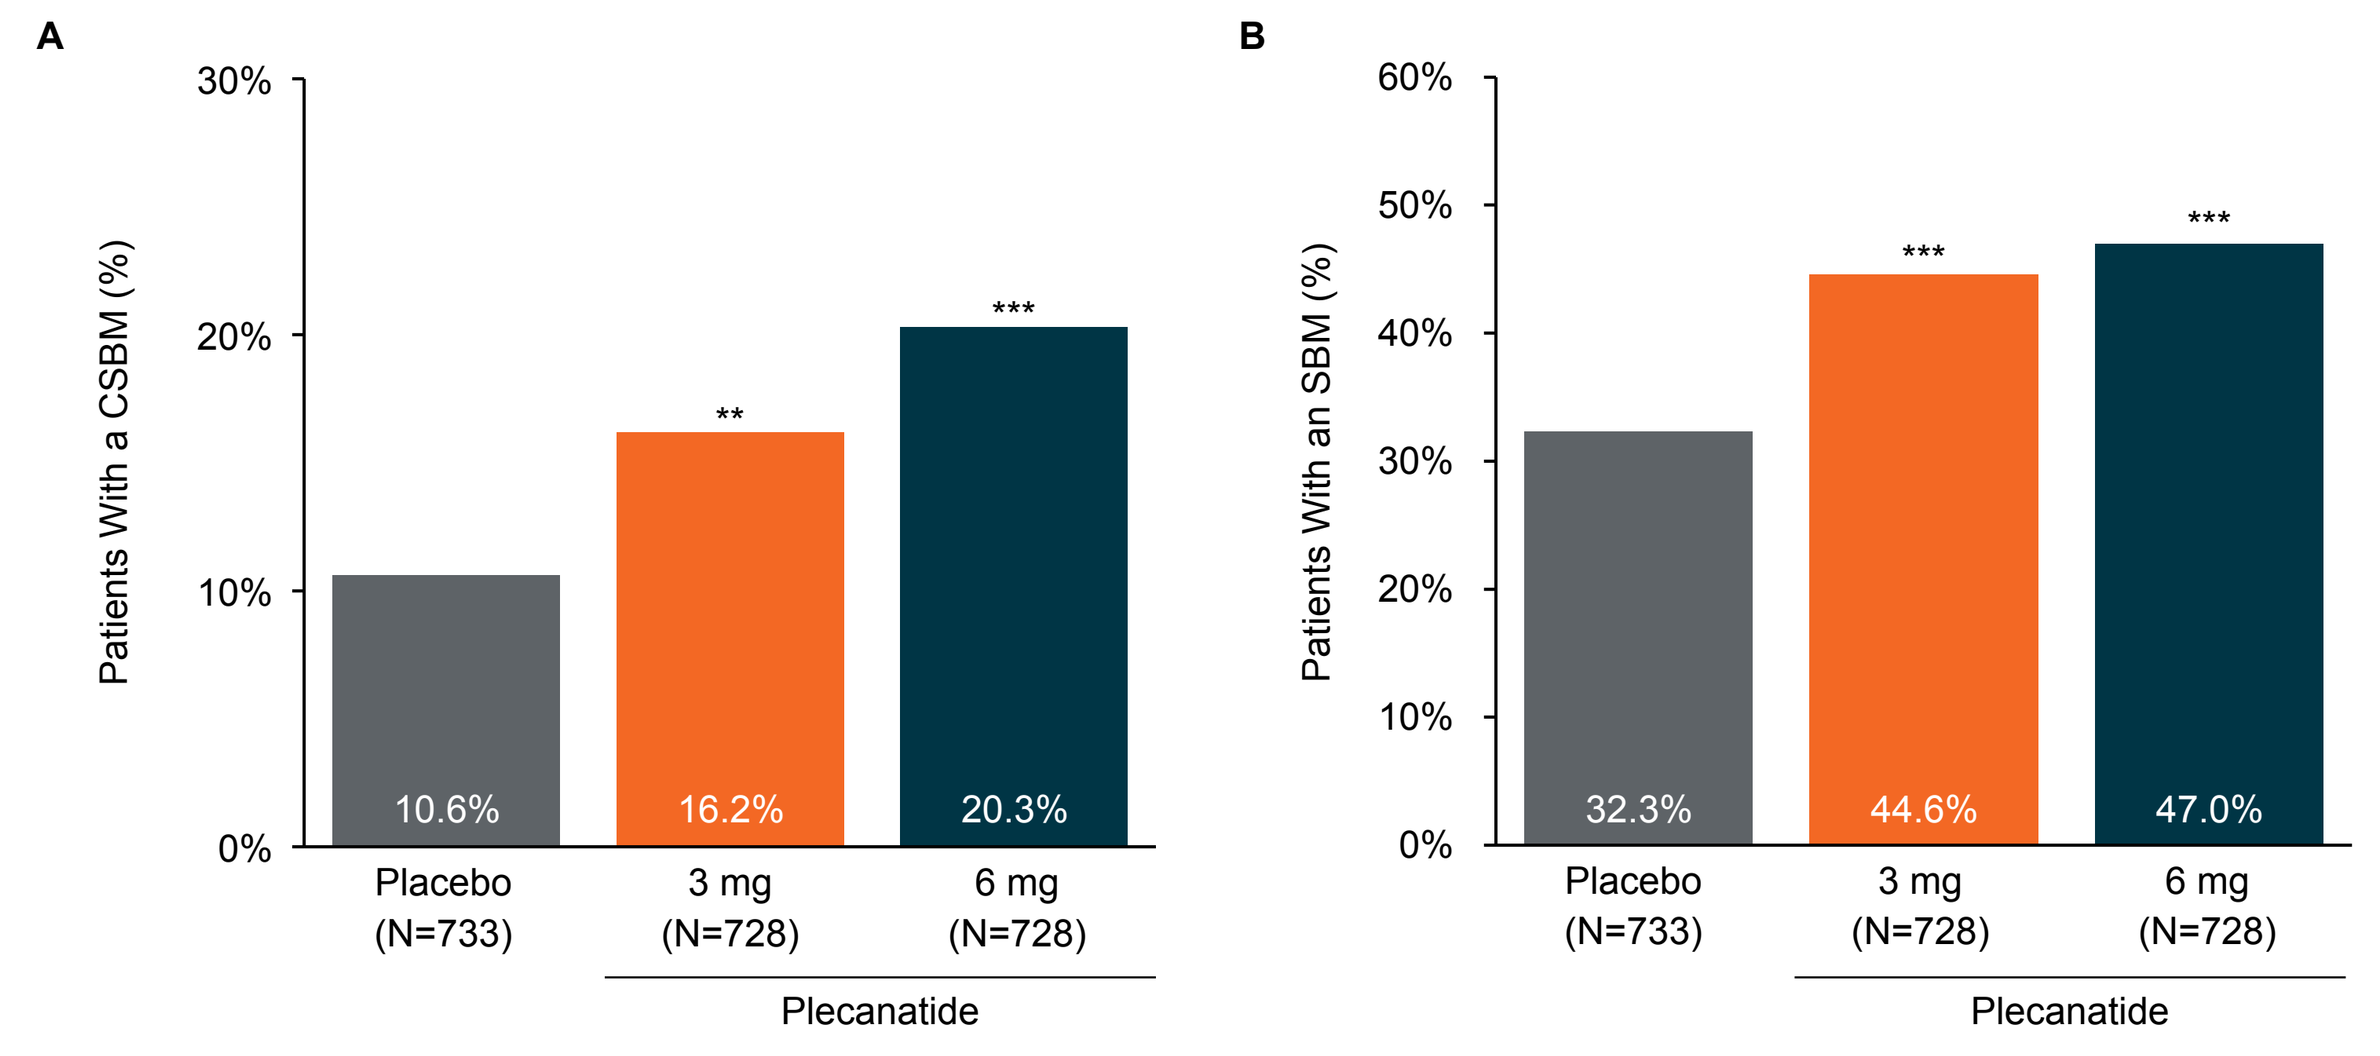
A
Patients With a CSBM (%)
30%
20%
10%
0%
**

10.6%
16.2%
20.3%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide
B
Patients With an SBM (%)
60%
50%
40%
30%
20%
10%
0%

32.3%
44.6%
47.0%
Placebo (N=733)
3 mg (N=728)
6 mg (N=728)
Plecanatide

P<0.01, *P<0.001 vs placebo. CSBM, complete spontaneous bowel movement; SBM, spontaneous bowel movement.

- Significantly more plecanatide-treated patients experienced a CSBM (Figure 6A) or an SBM (Figure 6B) within 24 hours after the first dose of study medication compared to placebo-treated patients.

Table 2. Change From Baseline in Secondary Endpoints[a]

	Placebo (N=733)	Plecanatide 3 mg (N=728)	Plecanatide 6 mg (N=728)
CSBMs/week, LS mean (SE)	0.74 (0.078)	1.22 (0.079)***	1.41 (0.079)***
Stool consistency, LS mean (SE)[b]	0.90 (0.055)	1.42 (0.054)***	1.48 (0.055)***
Straining severity, LS mean (SE)[c]	–1.41 (0.080)	–2.02 (0.078)***	–2.11 (0.080)***
Abdominal pain, LS mean (SE)[c]	–1.26 (0.070)	–1.57 (0.069)***	–1.63 (0.070)***
Abdominal bloating, LS mean (SE)[c]	–1.19 (0.068)	–1.51 (0.068)***	–1.56 (0.068)***
Treatment satisfaction, mean[d]	2.4	2.8***	2.7***
Treatment continuation, mean[d]	3.4	3.8***	3.8***

***P<0.001 vs placebo.
[a]LS mean values are the overall average estimate across the 12-week treatment period. [b]Measured using the 7-point Bristol Stool Form Scale. [c]Measured using an 11-point scale, where 0=none and 10=worst possible. [d]Mean absolute score at week 12; measured using a 5-point scale where 1=not at all satisfied/likely and 5=very satisfied/likely. CSBM, complete spontaneous bowel movement; LS, least squares; SE, standard error.

- Plecanatide significantly improved the frequency of CSBMs, as well as patient-reported symptoms, including stool consistency, straining severity, and abdominal pain and bloating (Table 2).
- At week 12, significantly more plecanatide-treated patients indicated being satisfied with and having a desire to continue with treatment compared to placebo-treated patients (P<0.001, all comparisons).

Table 3. Summary of Adverse Events (AEs)

Patients, n (%)	Placebo (N=730)	Plecanatide 3 mg (N=726)	Plecanatide 6 mg (N=726)
≥1 AE	136 (18.6%)	173 (23.8%)	144 (19.8%)
Diarrhea	7 (1.0%)	31 (4.3%)	29 (4.0%)
AE by maximum severity			
Mild	85 (11.6%)	96 (13.2%)	78 (10.7%)
Moderate	44 (6.0%)	60 (8.3%)	55 (7.6%)
Severe	7 (1.0%)	17 (2.3%)	11 (1.5%)
AE leading to discontinuation	3 (0.4%)	18 (2.5%)	16 (2.2%)
Diarrhea	0	9 (1.2%)	10 (1.4%)
Serious AEs	6 (0.8%)	6 (0.8%)	5 (0.7%)

Two deaths were reported during the study. One patient succumbed to a pulmonary embolism during the screening period (ie, did not receive study drug). The second death was due to accidental drowning, which occurred after randomization, and was considered not related to study drug.

- The rate of AEs was similar across treatment groups (Table 3), with diarrhea as the only AE occurring in ≥2% of patients and at an incidence greater than placebo.
- Discontinuation due to an AE occurred in 2.3% of plecanatide-treated patients compared with 0.4% of placebo-treated patients, with diarrhea being the most common.
- Severe diarrhea was reported in 1.0% and 0.4% of patients in the plecanatide 3-mg and 6-mg treatment groups, respectively, and in 0.1% of patients receiving placebo.

DISCUSSION

- The hallmark symptoms of IBS-C (abdominal pain and infrequent stools), as well as secondary symptoms (stool consistency, straining severity, abdominal bloating), were significantly improved with 12 weeks of plecanatide treatment compared with placebo.
- Both components of the primary Overall Responder endpoint (reduction in abdominal pain plus increase in weekly CSBM frequency) demonstrated statistically significant results; thus, both contributed to the significance of the primary endpoint.
- Plecanatide-treated patients experienced low rates of AEs, including diarrhea, and low rates of treatment discontinuation due to diarrhea, indicating a benign safety and tolerability profile.
- Results from the Patient Global Assessment questionnaires indicated a greater overall satisfaction with plecanatide compared with placebo and a greater intention to continue treatment in patients receiving plecanatide than in those receiving placebo.
- The pooled results of two large-scale, randomized, double-blind, placebo-controlled, phase 3 trials of adults with IBS-C demonstrated that once-daily oral plecanatide (at 3 mg and 6 mg) offers a promising new treatment option for patients with IBS-C.

References

1. Hungin AP, Chang L, Locke GR, et al. *Aliment Pharmacol Ther*. 2005;21(11):1365-75.
2. Lacy BE, Mearin F, Chang L, et al. *Gastroenterology*. 2016;150(6):1393-407.e5.
3. Heidelbaugh JJ, Stelwagon M, Miller SA, et al. *Am J Gastroenterol*. 2015;110(4):580-7.
4. Ruiz-López MC, Coss-Adame E. *Rev Gastroenterol Mex*. 2015;80(1):13-20.
5. Nyrop KA, Palsson OS, Levy RL, et al. *Aliment Pharmacol Ther*. 2007;26(2):237-48.
6. Rey E, Balboa A, Mearin F. *Am J Gastroenterol*. 2014;109(6):876-84.
7. Sharma A, Herekar AA, Bhagatwala J, Rao SS. *Clin Exp Gastroenterol*. 2019:12:31-6.
8. Boulete IM, Thadi A, Beaufrand C, et al. *World J Gastroenterol*. 2018;24(17):1888-900.
9. Trulance (plecanatide) [package insert]. Bridgewater, NJ: Salix Pharmaceuticals Inc.; 2019.
10. Brenner DM, Fogel R, Dorn SD, et al. *Am J Gastroenterol*. 2018;113(5):735-45.

Disclosures: H. Franklin is an employee and stockholder at Salix Pharmaceuticals Inc.; J. Christie has nothing to disclose.
Acknowledgments: Funding for this study was provided by Synergy Pharmaceuticals Inc. Poster support was funded by Salix Pharmaceuticals Inc. Medical writing and editorial support was provided by The Medicine Group (New Hope, PA, USA), in accordance with Good Publication Practice guidelines.

Contact
Dr. Howard Franklin, Howard.Franklin@salix.com

This poster was funded by Salix Pharmaceuticals.

Salix Pharmaceuticals