Pharmacist Consultation in Individuals with Chronic Idiopathic Constipation or Irritable Bowel Syndrome with Constipation: Results from the BURDEN-CIC and BURDEN IBS-C Surveys

Eamonn M. M. Quigley, MD,[1] Kate Kim, PharmD,[2] Reema Patel, PharmD,[2] Lucinda A. Harris, MD[3]

[1]Houston Methodist Hospital, Houston, TX, USA; [2]Bausch Health, LLC., Bridgewater, NJ, USA; [3]Mayo Clinic Scottsdale, Scottsdale, AZ, USA

Scan here to obtain a PDF of this poster.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

88
Presented at the APhA 2020 Annual Meeting & Exposition; March 20–23, 2020; National Harbor, MD

BACKGROUND

- Symptoms of chronic idiopathic constipation (CIC) and irritable bowel syndrome with constipation (IBS-C) have a quantifiable negative impact on workplace and school productivity, as well as quality of life, with the overall goal of treatment being to relieve symptoms.[1-4]
- With the wide availability of over-the-counter treatments for these conditions, individuals with CIC and IBS-C are likely to self-treat their constipation and may not present to a healthcare provider.
 - The BURDEN IBS-C survey found that 76% of respondents were currently using a nonprescription treatment for their condition, including 71% of respondents who had never been formally diagnosed by a healthcare provider or healthcare extender.[4]
- Pharmacists can play a critical role in engaging with individuals with symptoms of CIC and IBS-C to address their concerns and optimize treatment.[5,6]
 - A study in Japan found that patients who consulted a pharmacist prior to purchasing nonprescription laxatives were more likely to use the appropriate medication and to identify and be aware of how to handle medication side effects.[7]
 - Though underutilized, individuals who consult with pharmacists tend to be satisfied with the consultation and more likely to reconsult with a pharmacist in the future.[8]
 - Pharmacists are positioned to refer these individuals to their general healthcare provider if they notice that the individual may be suffering from a more severe condition than can be relieved with over-the-counter treatments.[6]

OBJECTIVE

- The purpose of this analysis was to explore the use of pharmacists, in consideration of their position in the community setting, as medical consultants to patients with CIC and IBS-C using data from the BURDEN-CIC and BURDEN IBS-C surveys.

METHODS

- The BURDEN-CIC and BURDEN IBS-C surveys were designed to better understand the experiences, attitudes, and unmet needs of patients with CIC or IBS-C.[3,4]
- BURDEN-CIC and BURDEN IBS-C were institutional review board–approved online surveys that used proprietary databases to identify patients with CIC and IBS-C symptomatology, respectively.
 - Both surveys were conducted between June 29, 2016, and January 30, 2017.
- Patients were eligible to participate if they (A) self-reported a formal diagnosis of CIC or IBS-C, or (B) fulfilled Rome IV criteria for CIC or IBS-C as determined in the Screening Section of the survey.
 - Respondents were not eligible to participate if they had IBS with diarrhea, mixed IBS, undefined IBS, inflammatory bowel disease, diverticulitis, diverticulosis, spastic colon, celiac disease, cancer of the gastrointestinal tract, or if they had regularly taken an opioid (narcotic) within the past 3 months.
- Qualifying panelists completed an online, self-administered questionnaire, with answer types including dichotomous, multiple-choice, and open-ended formats, as well as Likert rating scales (1–5 or 1–7).
- The Patient Questionnaire sample was normalized (weighted) to correct for any biases in sampling or non-response using demographic and geographic distributions from the Current Population Survey as benchmarks.[9]
- Although pharmacists were not a target healthcare provider of either survey, here we focus on questions related to pharmacists and pharmacy-based care.

RESULTS

Table 1. Demographics of Participating Respondents

	BURDEN-CIC N=1223	BURDEN IBS-C N=1681
Age, years, mean (SD)	49.1 (18.0)	46.2 (15.9)
Age at Symptom Onset, years, mean (SD)	44.9 (17.7)	43.8 (16.1)
Diagnosed by an HCP, %	85%	62%
Sex, female, %	69%	73%
Race, %		
White / non-Hispanic	65%	65%
Black / non-Hispanic	13%	11%
Mixed / non-Hispanic	3%	5%
Other / non-Hispanic	3%	4%
Hispanic	16%	16%
Education, %		
High school or less	34%	36%
Some college	26%	36%
Bachelor's degree or higher	40%	28%

Note that percentages may not total 100% due to rounding. HCP, healthcare provider; SD, standard deviation.

- A total of 1223 participants with CIC and 1681 participants with IBS-C completed the survey.
- Demographics were similar between studies, with the exception that more participants with CIC had been formally diagnosed by a healthcare provider than had participants with IBS-C (**Table 1**).

Figure 1. Word Cloud Describing Meaning of Constipation for Participants with CIC

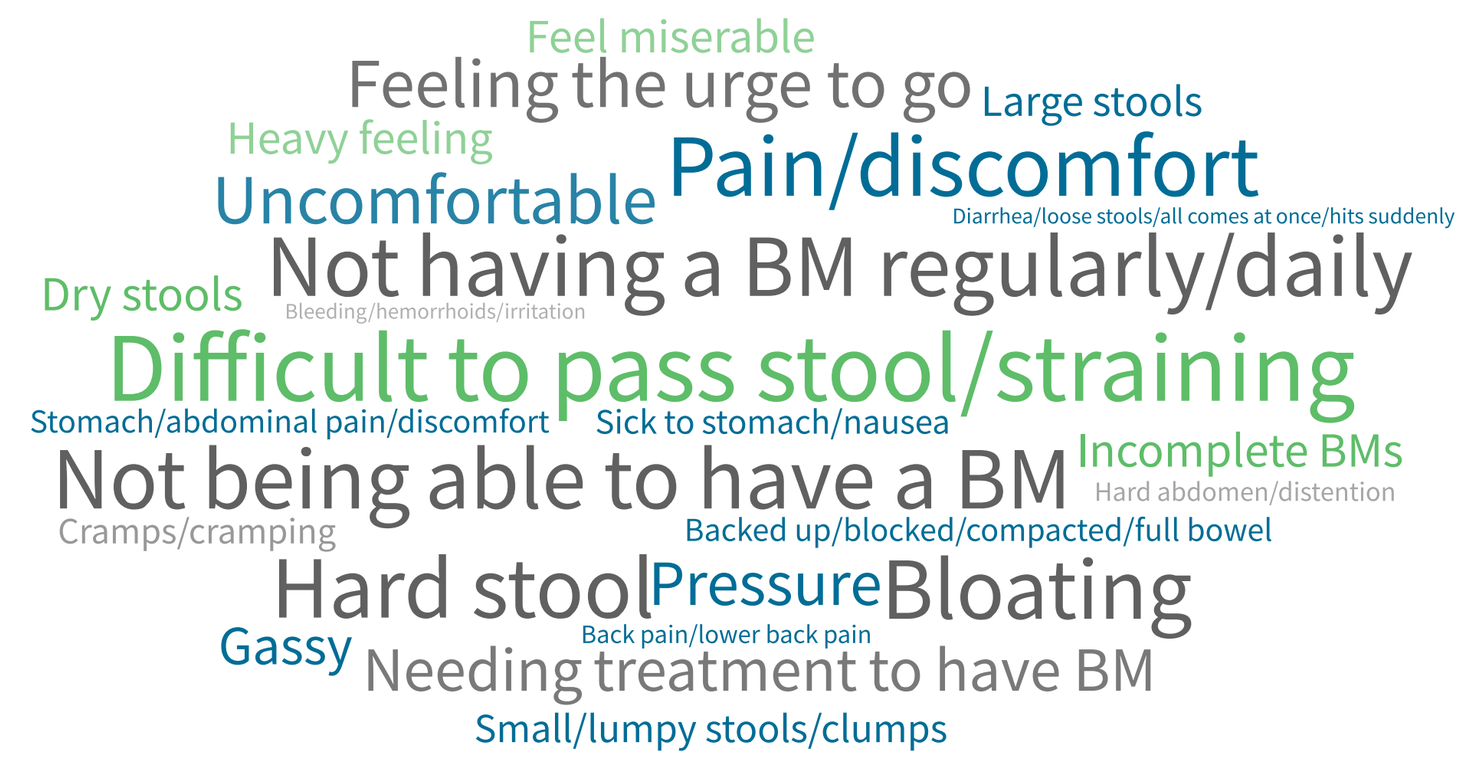
Feel miserable
Feeling the urge to go
Large stools
Heavy feeling
Uncomfortable
Pain/discomfort
Diarrhea/loose stools/all comes at once/hits suddenly
Dry stools
Not having a BM regularly/daily
Bleeding/hemorrhoids/irritation
Difficult to pass stool/straining
Stomach/abdominal pain/discomfort
Sick to stomach/nausea
Not being able to have a BM
Incomplete BMs
Hard abdomen/distention
Cramps/cramping
Backed up/blocked/compacted/full bowel
Hard stool
Pressure
Bloating
Back pain/lower back pain
Gassy
Needing treatment to have BM
Small/lumpy stools/clumps

BM, bowel movement.

- When asked to describe what constipation means to them (free-text), participants with CIC most often used phrases around "difficult to pass stool/straining" (35%), "not having a bowel movement regularly/daily" (30%), and "not being able to have a bowel movement" (26%) (**Figure 1**).
- Having a bowel movement "infrequently" was most commonly defined as once in 3 days (37%), followed by once in 4–5 days (22%) and once in 2 days (20%).

Figure 2. Word Cloud Describing Meaning of Constipation for Participants with IBS-C

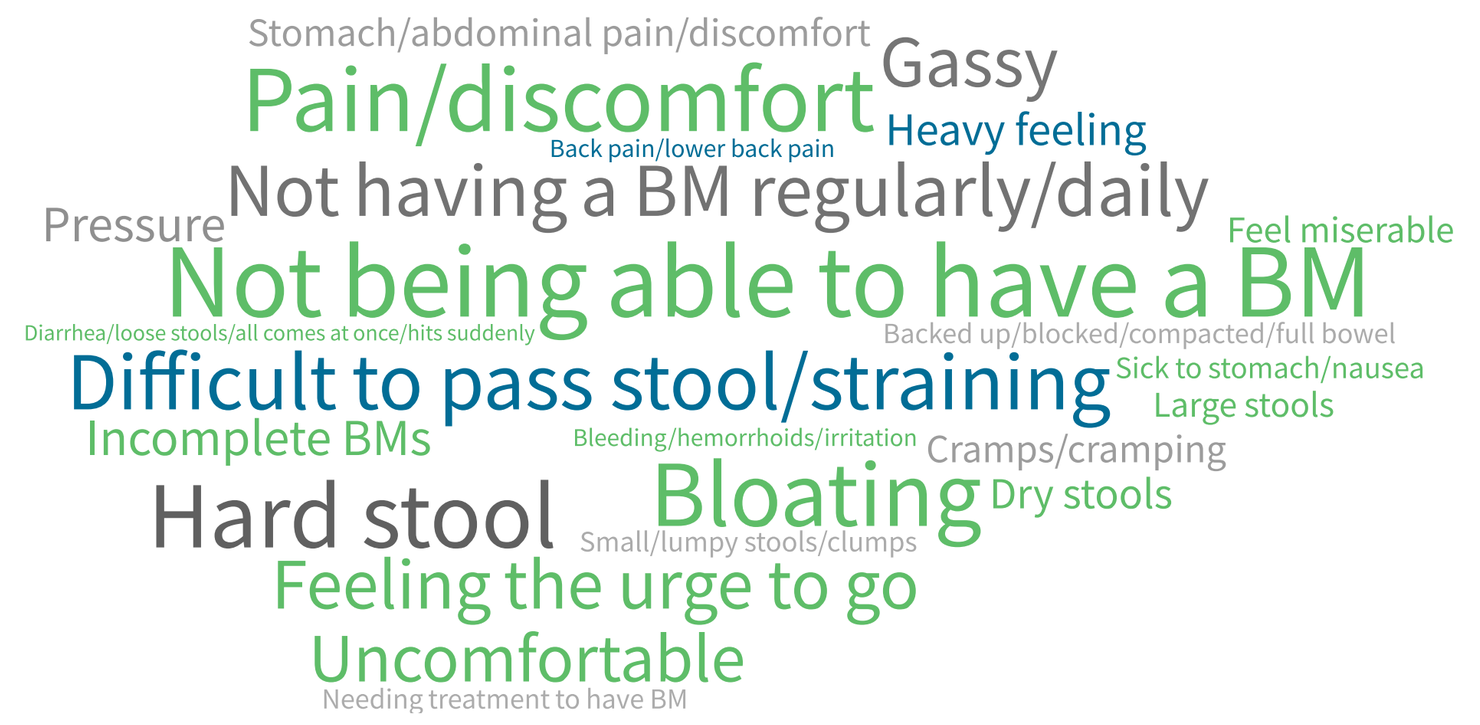
Stomach/abdominal pain/discomfort
Pain/discomfort
Gassy
Back pain/lower back pain
Heavy feeling
Pressure
Not having a BM regularly/daily
Feel miserable
Not being able to have a BM
Diarrhea/loose stools/all comes at once/hits suddenly
Backed up/blocked/compacted/full bowel
Difficult to pass stool/straining
Sick to stomach/nausea
Large stools
Incomplete BMs
Bleeding/hemorrhoids/irritation
Cramps/cramping
Hard stool
Bloating
Dry stools
Small/lumpy stools/clumps
Feeling the urge to go
Uncomfortable
Needing treatment to have BM

BM, bowel movement.

- Similar to participants with CIC, participants with IBS-C most often used phrases around "not being able to have a bowel movement" (40%), "difficult to pass stool/straining" (30%), and "not having a bowel movement regularly/daily" (26%) when asked to describe what constipation means to them (free-text) (**Figure 2**).
- Having a bowel movement "infrequently" was most commonly defined as once in 3 days (36%), followed by once in 2 days (29%) and once in 4–5 days (17%).

Figure 3. Participants Who Consulted with Their Pharmacist by Formal Diagnosis

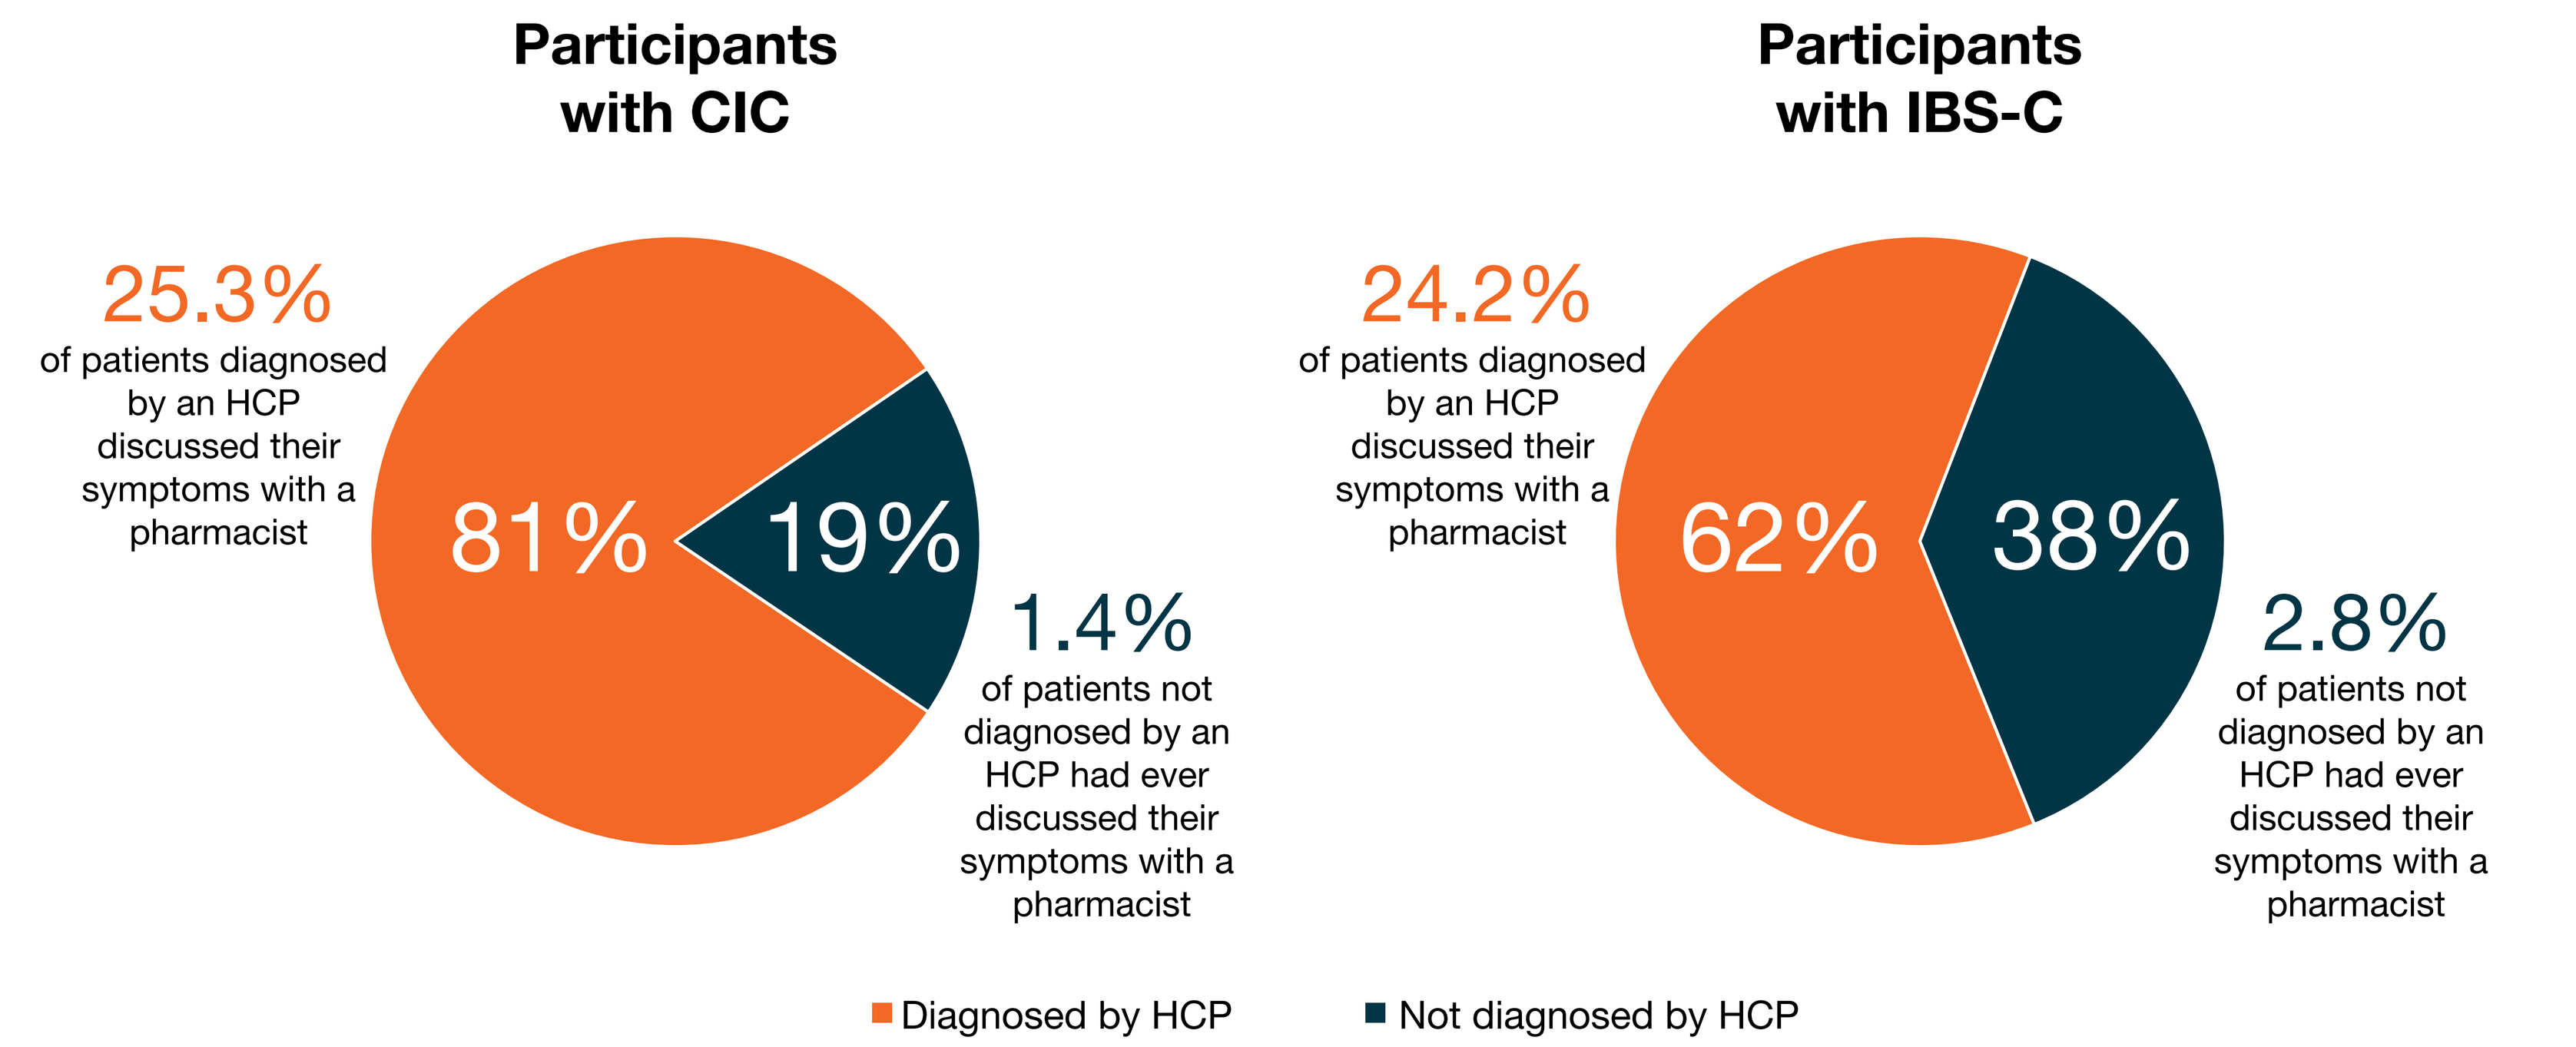
Participants with CIC
25.3%
of patients diagnosed by an HCP discussed their symptoms with a pharmacist
81%
19%
1.4%
of patients not diagnosed by an HCP had ever discussed their symptoms with a pharmacist
Participants with IBS-C
24.2%
of patients diagnosed by an HCP discussed their symptoms with a pharmacist
62%
38%
2.8%
of patients not diagnosed by an HCP had ever discussed their symptoms with a pharmacist
Diagnosed by HCP
Not diagnosed by HCP

HCP, healthcare provider.

- Most patients in both surveys had been formally diagnosed with CIC or IBS-C by a healthcare provider, with approximately one-fourth of those participants having consulted a pharmacist; very few undiagnosed participants had ever spoken to a pharmacist regarding their symptoms (**Figure 3**).
- Diagnosed participants with IBS-C were more likely to discuss symptoms with a pharmacist after consulting a healthcare provider (ie, medical doctor, physicians' assistant, nurse, or nurse practitioner [before, 10.4%; after, 13.9%).
- Participants with diagnosed CIC and undiagnosed participants with IBS-C were equally likely to consult with a pharmacist before or after consulting a healthcare provider.

Figure 4. Treatments Tried Prior to Consulting a Healthcare Provider

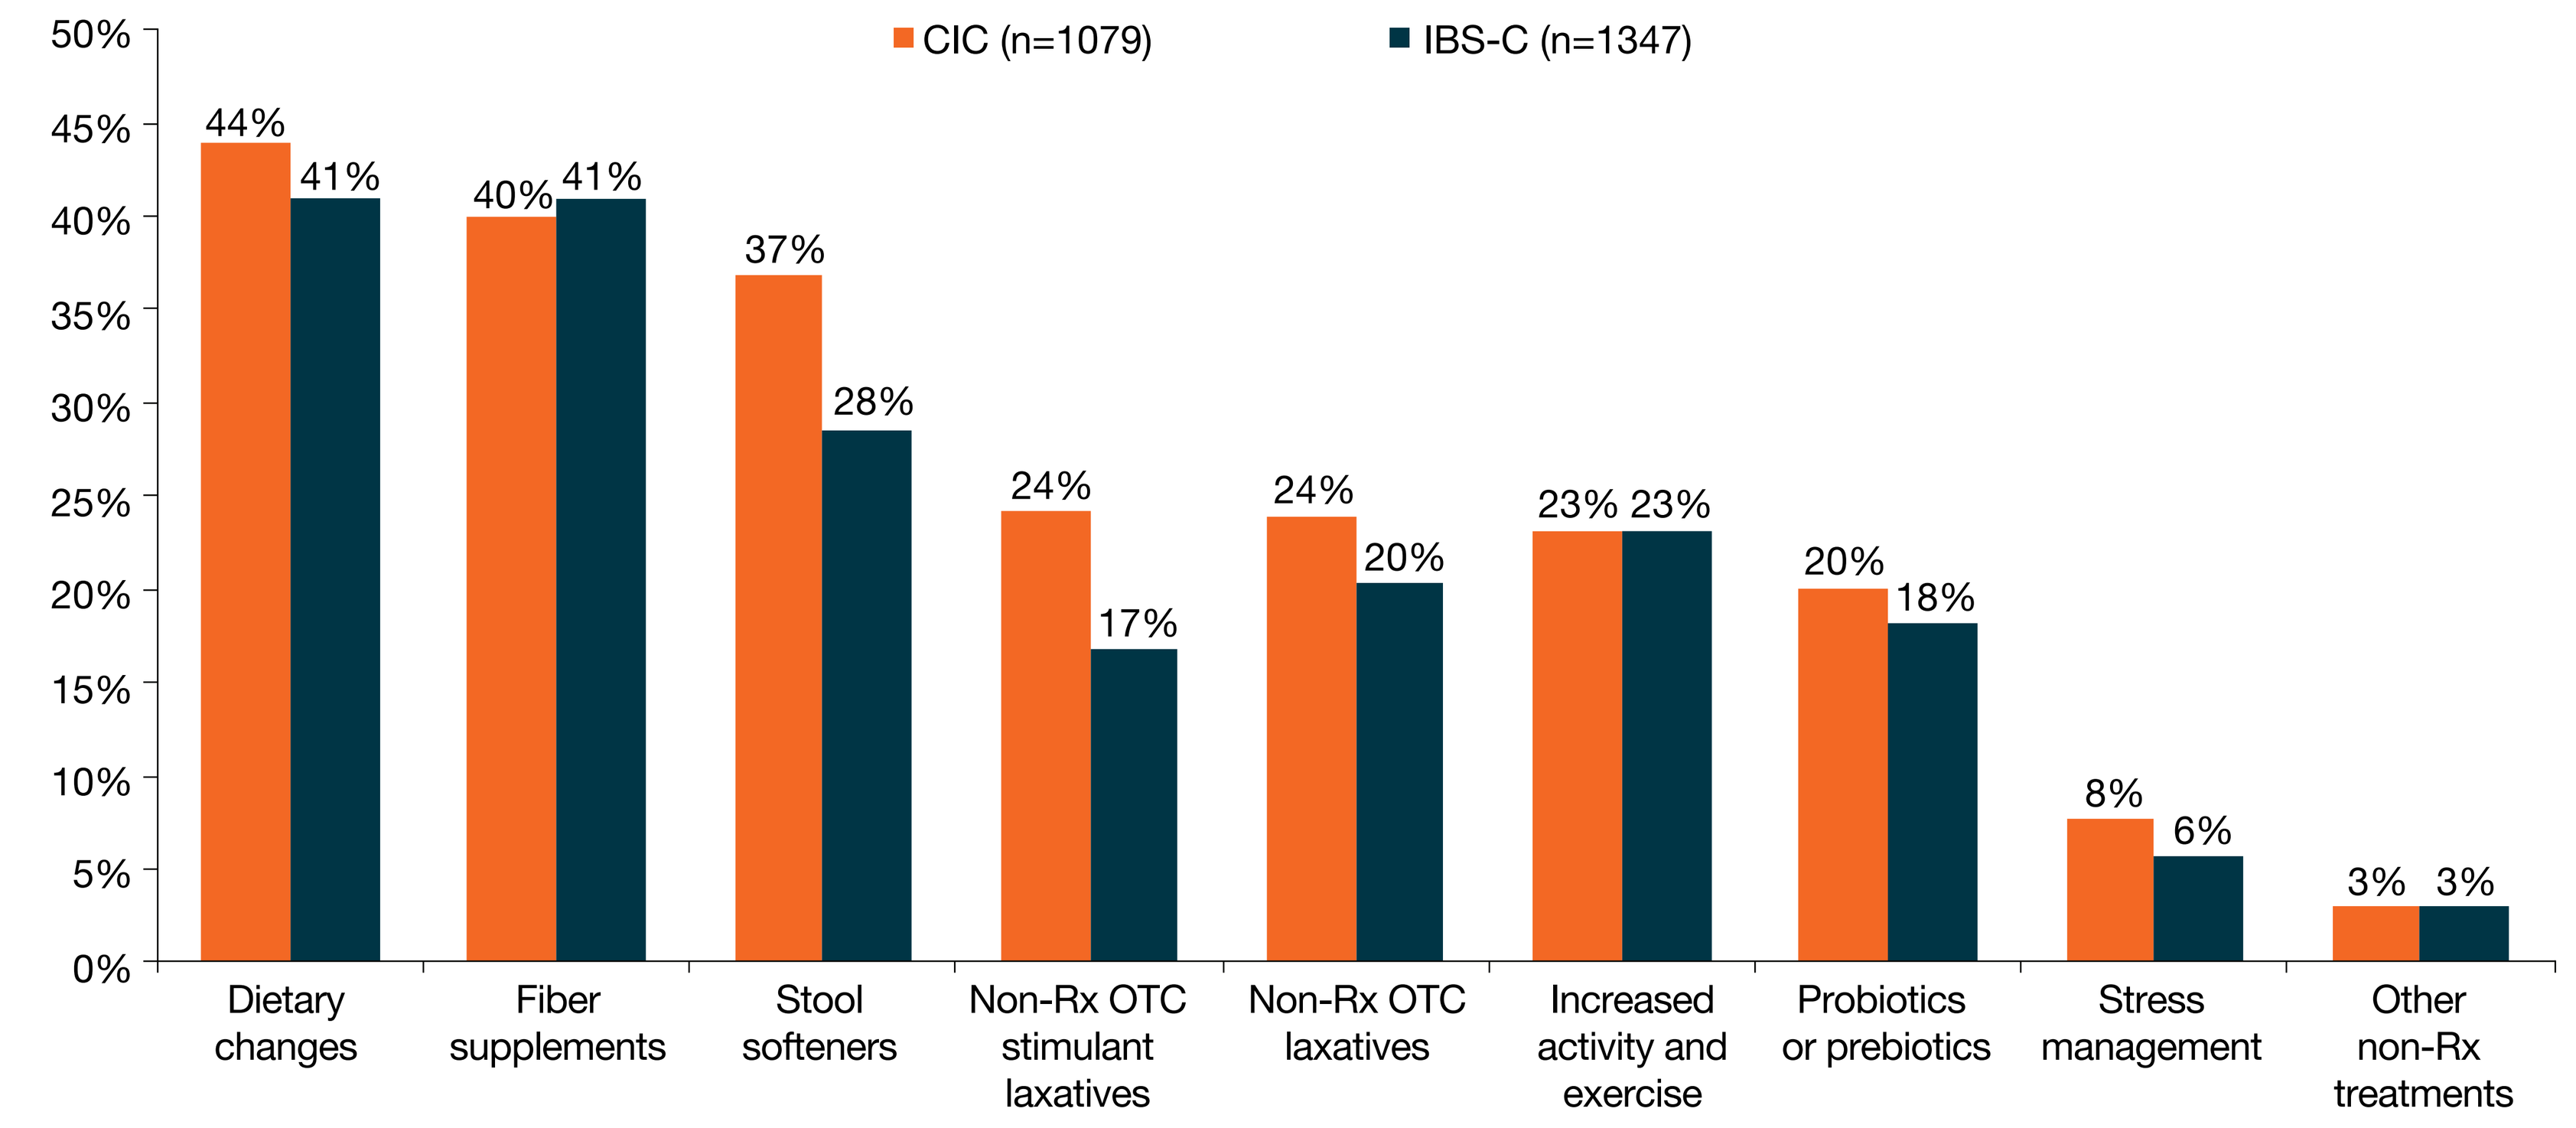
CIC (n=1079)
IBS-C (n=1347)
50%
45%
40%
35%
30%
25%
20%
15%
10%
5%
0%
44%
41%
40%
41%
37%
28%
24%
17%
24%
20%
23% 23%
20%
18%
8%
6%
3% 3%
Dietary changes
Fiber supplements
Stool softeners
Non-Rx OTC stimulant laxatives
Non-Rx OTC laxatives
Increased activity and exercise
Probiotics or prebiotics
Stress management
Other non-Rx treatments

OTC, over the counter; Rx, prescription.

- Among participants who had ever discussed their symptoms with any healthcare provider (including pharmacists) and had tried to manage their symptoms, many had attempted various methods of treating their symptoms—the most common being dietary changes and fiber supplements (**Figure 4**).
- On average, these participants had tried 3.4 (CIC) and 3.7 (IBS-C) over-the-counter products prior to speaking with an HCP.

DISCUSSION / CONCLUSIONS

- Results from BURDEN-CIC and BURDEN IBS-C reveal that individuals with CIC or IBS-C who were not formally diagnosed are not using pharmacists as a source of information for managing their symptoms, even though nearly half of individuals with CIC or IBS-C are using over-the-counter medications to treat their symptoms before consulting a medical doctor / physician assistant / nurse practitioner.

References

1. Cash BD. *Gastroenterol Hepatol (NY)*. 2018;14(5 Suppl 3):3-15.
2. Heidelbaugh JJ, Stelwagon M, Miller SA, Shea EP, Chey WD. *Am J Gastroenterol*. 2015;110(4):580-587.
3. Harris LA, Horn J, Kissous-Hunt M, Magnus L, Quigley EMM. *Adv Ther.* 2017;34(12):2661-2673.
4. Quigley EMM, Horn J, Kissous-Hunt M, Crozier RA, Harris LA. *Adv Ther.* 2018;35(7):967-980.
5. Bell J, Dziekan G, Pollack C, Mahachai V. *Adv Ther.* 2016;33(10):1691-1703.
6. Gilson AM, Stone JA, Reddy A, Chui MA. *J Am Pharm Assoc (2003)*. 2019;59(6):852-856.
7. Shibata K, Matsumoto A, Nakagawa A, et al. *Biol Pharm Bull*. 2016;39(11):1767-1773.
8. Schimmelfing JT, Brookhart AL, Fountain KMB, Goode JKR. *J Am Pharm Assoc (2003)*. 2017;57(1):86-89.e81.
9. Knowledge Panel® Recruitment and Sample Survey Methodologies. (5/25/17). http://www.gfk.com/fileadmin/user_upload/dyna_content/US/documents/KnowledgePanel_Methodology.pdf. Accessed 1/15/2020.

Disclosures: E. M. M. Quigley is the David M Underwood Chair of Medicine in Digestive Disorders, Co-Director of the Lynda K and David M Underwood Center for Digestive Disorders, Chief of Gastroenterology and Hepatology, Professor of Medicine at the Institute of Academic Medicine, Houston Methodist Hospital; Professor of Medicine at the Weill Cornell Medical College; and Adjunct Professor of Medicine, Texas A&M Health Sciences Center College of Medicine.
K. Kim is an employee of Bausch Health.
R. Patel is an employee and stockholder at Bausch Health.
L. A. Harris is a consultant for Salix, Ironwood, Allergan, Takeda (Shire), and Rome Foundation.
Acknowledgment: Funding for this study and poster support was provided by Salix Pharmaceuticals Inc. Medical writing and editorial support was provided by The Medicine Group (New Hope, PA, USA), in accordance with Good Publication Practice guidelines.

This poster was funded by Salix Pharmaceuticals.