Efficacy of Plecanatide for Patients with Chronic Idiopathic Constipation and Irritable Bowel Syndrome–Constipation with Abdominal Bloating: Analysis from Four Randomized Phase 3 Clinical Trials

Darren Brenner, MD[1]; Howard Franklin, MD[2]; Gregory S. Sayuk, MD, MPH[3–5]

[1]Internal Medicine–Gastroenterology, Northwestern University–Feinberg School of Medicine, Chicago, IL, USA; [2]Salix Pharmaceuticals, Inc., Bridgewater, NJ, USA; [3]Division of Gastroenterology, Washington University School of Medicine, St. Louis, MO, USA; [4]Department of Psychiatry, Washington University School of Medicine, St. Louis, MO, USA; [5]Gastroenterology Section, John Cochran Veterans Affairs Medical Center, St. Louis, MO, USA

Scan here to obtain a PDF of this poster.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

P2157

Presented at the ACG Annual Scientific Meeting; October 25-30, 2019; San Antonio, TX

BACKGROUND

- Abdominal bloating is a key symptom of functional constipation disorders and is associated with reduced physical and mental quality of life,[1,2] and increased healthcare use compared to patients without bloating.[1]
- Bloating is common in patients with irritable bowel syndrome with constipation (IBS-C) and with chronic idiopathic constipation (CIC), although it is more prevalent and more severe in patients with IBS-C.[3]
- Bloating is also the second most commonly bothersome symptom of IBS-C, following abdominal cramping.[1]
- Plecanatide – approved for CIC and IBS-C – is a guanylate cyclase-C (GC-C) receptor agonist.[4] It is a structural analog of the naturally occurring human gastrointestinal (GI) peptide uroguanylin and replicates its pH-sensitive activity in preclinical studies.[5]
- Two CIC[6,7] and 2 IBS-C[8] trials with plecanatide have been completed and represent the largest phase 3 trials to date in each of these disorders.

AIM

- To provide an integrated analysis of the efficacy results obtained during the clinical development program for plecanatide for the treatment of CIC and IBS-C
- Pooled results from four phase 3 studies with a focus on abdominal bloating symptoms are presented herein

METHODS

- Each phase 3 trial was a 12-week, multicenter, randomized, double-blind, placebo-controlled clinical study.
- Safety populations from the 4 trials (2 CIC, 2 IBS-C) were combined for this post hoc analysis.
- Patients enrolled in these trials were adults (aged ≥18 years) who met modified Rome III criteria for either CIC or IBS-C.
- Upon completion of the 2-week baseline/screening period, patients were randomized to receive plecanatide 3 mg, plecanatide 6 mg, or placebo.
 - Medication was taken once daily with or without food.
- Efficacy assessments were made from the combined intent-to-treat (ITT) populations and were limited to endpoints that were common between the CIC and IBS-C trials.
- The primary endpoints for each study were:
 - The percentage of durable overall complete spontaneous bowel movement (CSBM) responders in the two studies performed in patients with CIC
 - Weekly CSBM responder: a patient who had ≥3 CSBMs/week and an increase from baseline of ≥1 CSBM for that week
 - Durable overall CSBM responder: a patient who was a weekly CSBM responder for ≥9 of the 12 treatment weeks, and at least 3 of the last 4 weeks of treatment
 - Overall responder, defined as a patient who was a weekly responder (CSBM + ≥30% improvement in abdominal pain) for ≥6 of the 12 treatment weeks, as defined previously for IBS-C
- Abdominal bloating was a secondary patient-reported endpoint in all studies.
 - In the CIC studies, abdominal bloating was reported on a 5-point Likert scale of 0 to 4 (0=none, 1=mild, 2=moderate, 3=severe, and 4=very severe).
 - In the IBS-C studies, abdominal bloating was reported on an 11-point Numeric Rating Scale from 0 (no abdominal bloating) to 10 (worst possible abdominal bloating).
- Daily bowel movements and symptoms were recorded in an electronic diary.
- Bloating outcomes were compared using pairwise comparisons of least-square means between the specified treatment group and the placebo group using a linear mixed-effects model with fixed effects for sex (stratification variable), baseline value, treatment, week, the interaction of treatment and week, and a random intercept for patient.
- Safety assessments were conducted using the collection of spontaneously reported adverse events (AEs) and responses to open-ended nonspecific questions.

RESULTS

Patients

- 2683 participants who met modified Rome III criteria for CIC and 2189 participants who met the criteria for IBS-C were included in combined phase 3 ITT–efficacy populations for each condition. Participants were predominantly female, white, and aged <65 years (Table 1).

Table 1. Patient Demographics and Baseline Characteristics (ITT Population)

	CIC			IBS-C		
		Plecanatide			Plecanatide	
N	**Placebo 897**	**3 mg 896**	**6 mg 890**	**Placebo 733**	**3 mg 728**	**6 mg 728**
Age, years, mean (SD)	45.5 (14.3)	45.2 (14.5)	45.2 (14.1)	43.9 (14.2)	43.5 (14.2)	43.5 (14.2)
Sex, n (%)						
Female	707 (78.8)	713 (79.6)	715 (80.3)	544 (74.2)	537 (73.8)	539 (74.0)
BMI, mean kg/m² (SD)	28.0 (5.2)	28.4 (5.0)	28.3 (5.1)	28.0 (4.8)	28.2 (4.8)	28.0 (4.9)
Ethnicity*, n (%)						
White	654 (72.9)	643 (71.8)	626 (70.3)	538 (73.4)	529 (72.7)	518 (71.2)
Black or African American	199 (22.2)	217 (24.2)	210 (23.6)	162 (22.1)	156 (21.4)	179 (24.6)
Other	44 (4.8)	36 (3.9)	54 (6.1)	30 (4.0)	37 (5.1)	29 (4.0)
Multiple	–	–	–	3 (0.4)	6 (0.8)	2 (0.3)
Weekly baseline values, mean (SD)						
CSBMs	0.35 (0.53)	0.31 (0.55)	0.28 (0.48)	0.23 (0.45)	0.24 (0.50)	0.27 (0.53)
SBMs	1.86 (1.85)	1.89 (1.92)	1.71 (1.74)	1.42 (1.09)	1.48 (1.08)	1.48 (1.14)
Abdominal bloating[a]	1.96 (0.91)	1.95 (0.92)	1.99 (0.94)	6.47 (1.77)	6.48 (1.70)	6.40 (1.78)
Patient global rating – IBS disease severity	–	–	–	3.6 (0.72)	3.6 (0.74)	3.5 (0.74)

*Ethnicity was self-reported. Percentages may not add up to 100% due to rounding.
[a]For CIC studies, abdominal bloating was assessed on a 5-point Likert scale where 0=none and 4=very severe; for IBS-C studies, abdominal bloating was assessed on an 11-point Numeric Rating Scale from 0 (no abdominal bloating) to 10 (worst possible abdominal bloating).
BMI, body mass index; CIC, chronic idiopathic constipation; CSBM, complete spontaneous bowel movement; IBS-C, irritable bowel syndrome with constipation-predominant symptoms; SBM, spontaneous bowel movement; SD, standard deviation.

Clinical Efficacy

Participants with CIC

- A significantly greater proportion of patients treated with plecanatide were classified as overall responders compared with those receiving placebo, (20.5% and 11.5%, respectively (*P*<0.001)).
- Significant improvement in abdominal bloating from baseline, compared with placebo, was observed in both the 3 and 6 mg plecanatide groups weeks 1 through 12 (**Figure 1A**).
- Plecanatide 3 and 6 mg significantly improved abdominal bloating scores by –0.12 points (95% CI, –0.18 to –0.06; *P*<0.001) and –0.08 points (95% CI, –0.14 to –0.02; *P*=0.009), respectively, after 12 weeks of treatment (**Figure 2**).
- Two weeks after ceasing treatment, no significant difference in abdominal bloating was observed between placebo and either treatment group.
- No worsening, compared with baseline, was reported at any timepoint.

Participants with IBS-C

- A significantly greater proportion of patients treated with plecanatide (3 or 6 mg) were classified as overall responders compared with those receiving placebo (25.7% and 16%, respectively (*P*<0.001)).
- Significant improvement in abdominal bloating from baseline, compared with placebo, was observed in both the 3 and 6 mg plecanatide groups after 1 week and persisted throughout 12 weeks of treatment (**Figure 1B**).
 - Change in abdominal bloating scores remained significantly lower 2 weeks after ceasing treatment when compared with placebo.
- Plecanatide 3 and 6 mg significantly improved abdominal bloating scores by –0.31 points (95% CI, –0.49 to –0.13; *P*<0.001) and –0.37 points (95% CI, –0.55 to –0.19; *P*<0.001), respectively, after 12 weeks of treatment (**Figure 2**).
- No worsening, compared with baseline, was reported at any timepoint, including 2 weeks after ceasing treatment.

Figure 1. Plecanatide Significantly Reduced Abdominal Bloating Severity Weeks 1-14

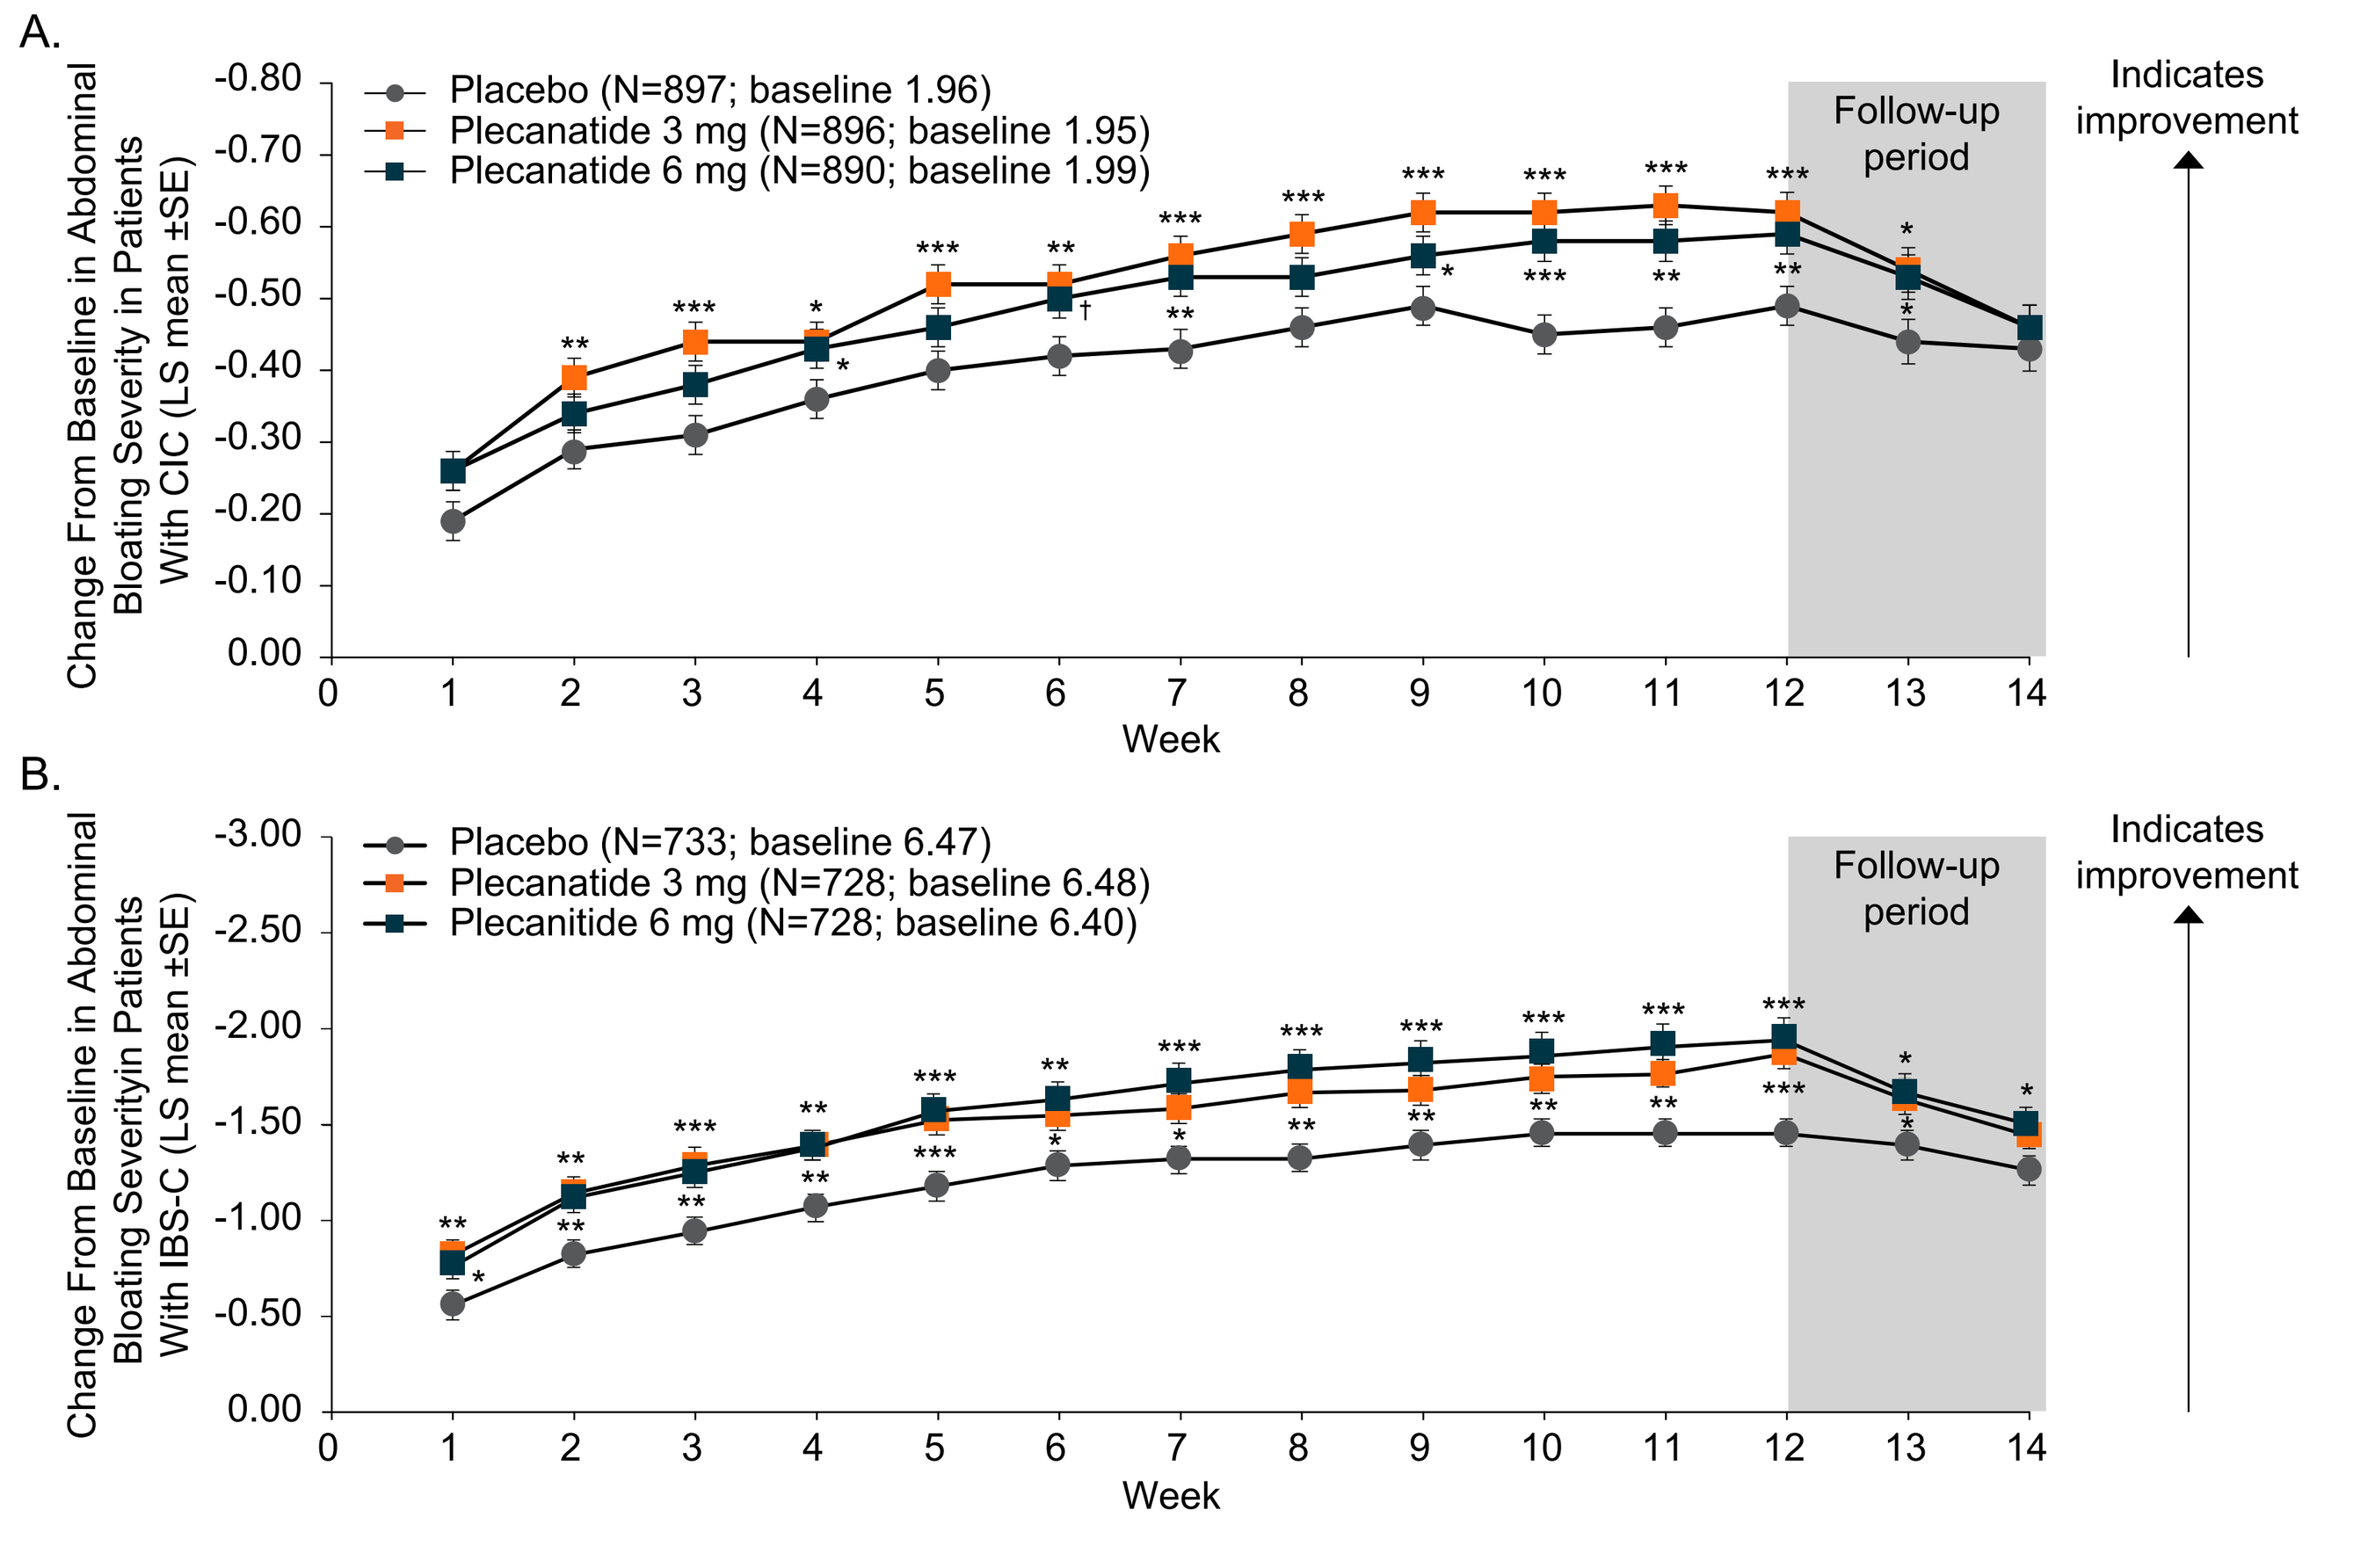
A.
Change From Baseline in Abdominal Bloating Severity in Patients With CIC (LS mean ±SE)
Placebo (N=897; baseline 1.96)
Plecanatide 3 mg (N=896; baseline 1.95)
Plecanatide 6 mg (N=890; baseline 1.99)
Follow-up period
Indicates improvement
Week
B.
Change From Baseline in Abdominal Bloating Severityin Patients With IBS-C (LS mean ±SE)
Placebo (N=733; baseline 6.47)
Plecanatide 3 mg (N=728; baseline 6.48)
Plecanitide 6 mg (N=728; baseline 6.40)
Follow-up period
Indicates improvement
Week

***P<0.001, **P<0.01, *P<0.05, †P=0.05.
LS=least squares; SE standard error.

Figure 2. Change in Abdominal Bloating Score in Patients With CIC (A) and IBS-C (B) Administered Placebo or Plecanatide (ITT Efficacy Population)

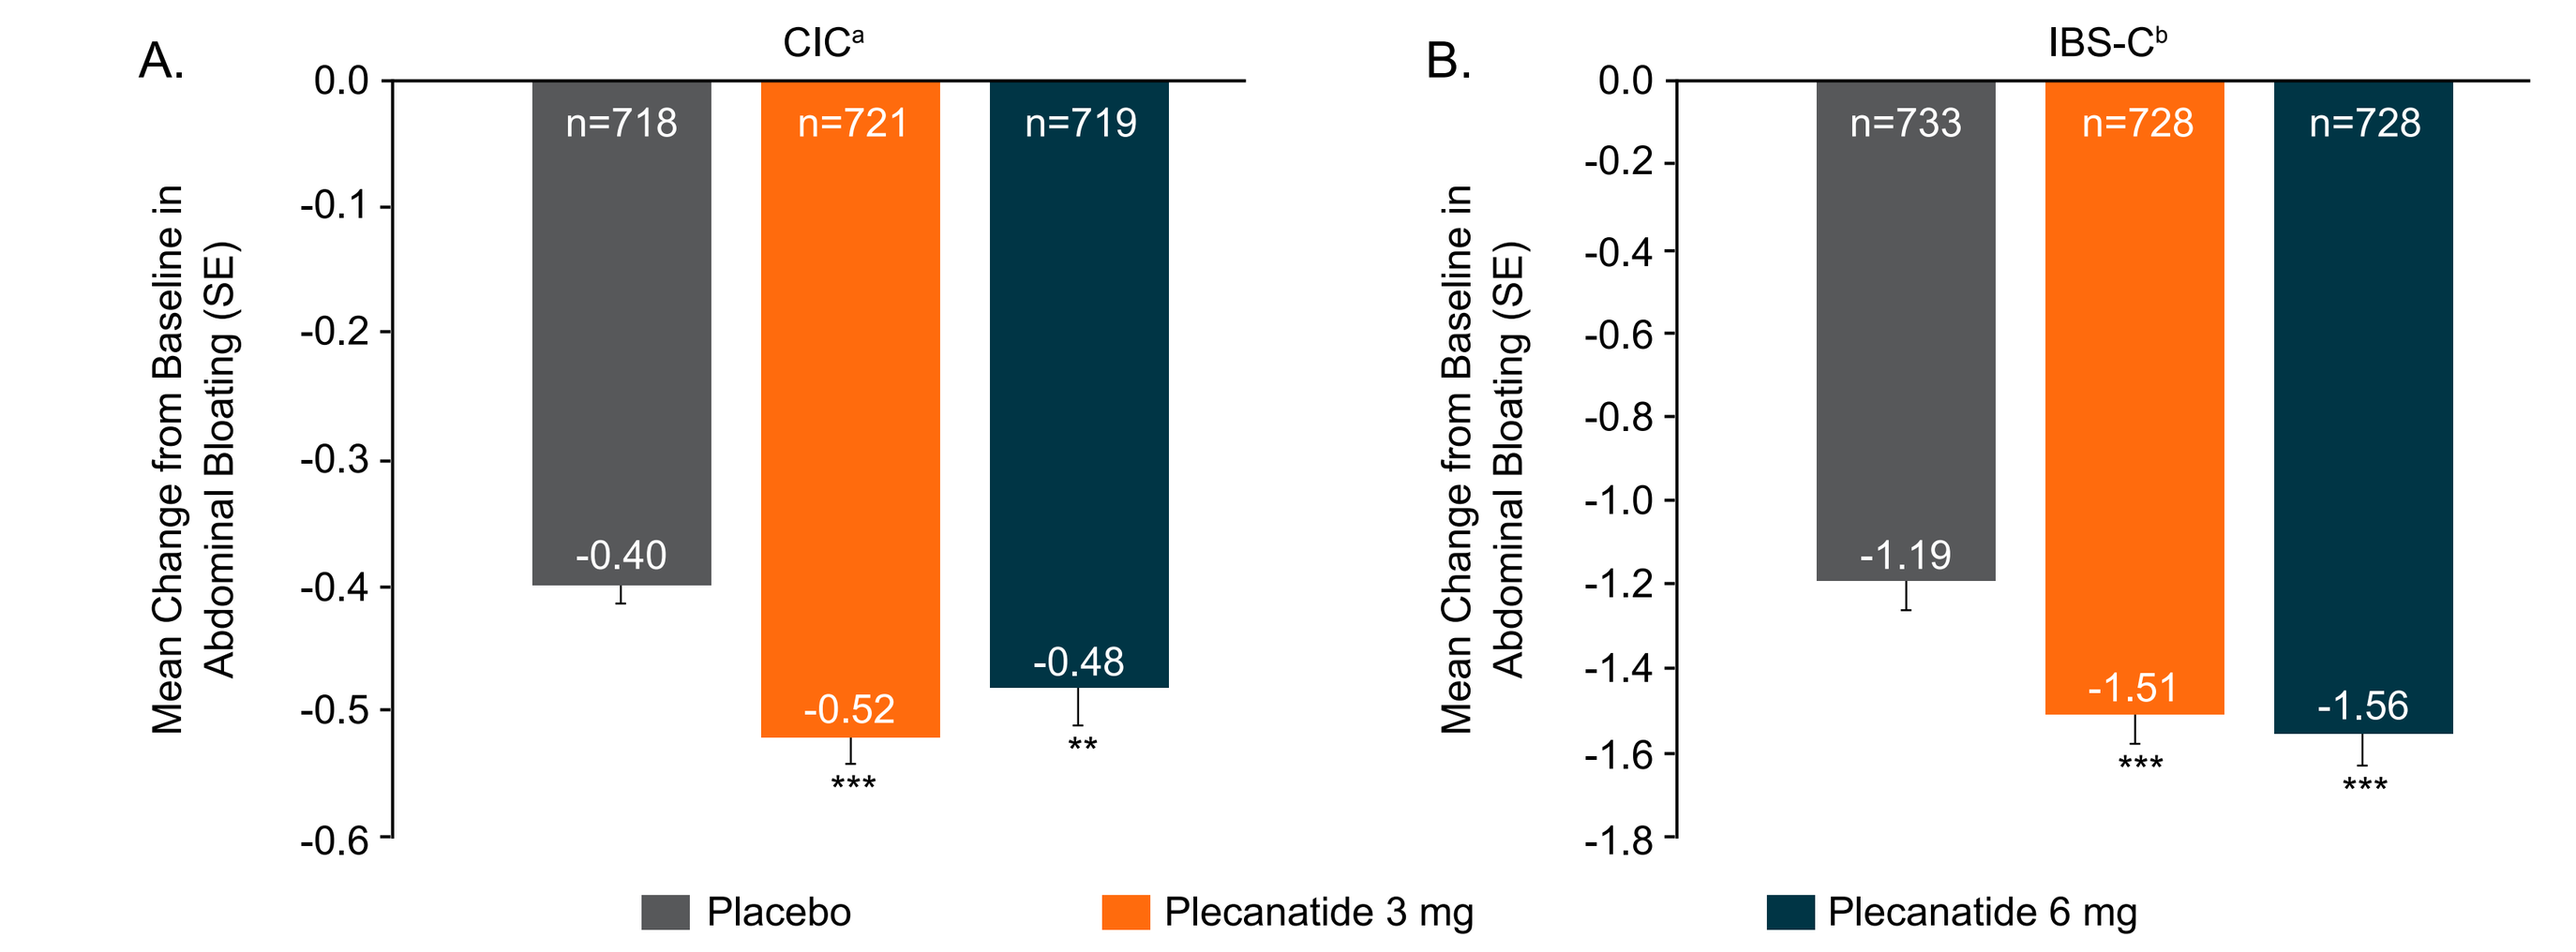
A.
CIC[a]
Mean Change from Baseline in Abdominal Bloating (SE)
n=718
n=721
n=719
-0.40
-0.52
-0.48
B.
IBS-C[b]
Mean Change from Baseline in Abdominal Bloating (SE)
n=733
n=728
n=728
-1.19
-1.51
-1.56
Placebo
Plecanatide 3 mg
Plecanatide 6 mg

P=0.009 versus placebo; *P<0.001 versus placebo.
[a]For CIC studies, abdominal bloating was assessed on a 5-point Likert scale where 0=none and 4=very severe.
[b]For IBS-C studies, abdominal bloating was assessed on an 11-point Numeric Rating Scale from 0 (no abdominal bloating) to 10 (worst possible abdominal bloating).
CIC, chronic idiopathic constipation; IBS-C, irritable bowel syndrome with constipation-predominant symptoms; ITT, intent-to-treat; SE, standard error of the mean.

Table 2. Summary of Adverse Events (Safety Population)

Patients, %		Placebo	Plecanatide 3 mg	Plecanatide 6 mg
CIC N=2791	Diarrhea	1.3%	4.6%	5.1%
	Discontinuation due to AEs	2.2%	4.1%	4.5%
	Discontinuation due to diarrhea	0.4%	1.9%	1.8%
IBS-C N=2182	Diarrhea	1.0%	4.3%	4.0%
	Discontinuation due to AEs	0.4%	2.5%	2.2%
	Discontinuation due to diarrhea	0	1.2%	1.4%

AE, adverse event; CIC, chronic idiopathic constipation; IBS-C, irritable bowel syndrome with constipation-predominant symptoms.

Safety and Tolerability

- The most common AE across all studies was diarrhea.
- The AE profiles for the 3-mg and 6-mg plecanatide dose groups were generally similar, except for a slight increase in severe diarrhea in the 6-mg dose group compared with the 3-mg dose group in patients with CIC.
- Discontinuation rates due to AEs, including diarrhea, were similar across studies and indications (**Table 2**).

CONCLUSIONS

- The efficacy of plecanatide for patients with both CIC and IBS-C is consistent with the concept of these disorders existing on a spectrum of functional constipation disorders.
- This integrated analysis of data from phase 3 studies confirms the efficacy of plecanatide for patients with CIC and IBS-C, including significantly improved bloating symptoms compared with placebo.
- Future research is needed as to whether the efficacy of plecanatide in significantly reducing the severity of abdominal bloating in patients with CIC and IBS-C may improve patient quality of life and reduce healthcare resource utilization.
- Plecanatide-treated patients experienced low rates of AEs, including diarrhea, and low rates of treatment discontinuation due to diarrhea, indicating a benign safety and tolerability profile.

References

1. Ringel Y, Williams RE, Kalilani L, Cook SF. *Clin Gastroenterol Hepatol*. 2009;7(1):68-72; quiz 63.
2. Neri L, Iovino P; Laxative Inadequate Relief Survey (LIRS) Group. *Neurogastroenterol Motil*. 2016;28(4)581-591.
3. Shah ED, Almario CV, Spiegel BMR, Chey WD. *J Neurogastroenterol Motil*. 2018; 24(2):299-306.
4. Trulance (plecanatide) [package insert]. Bridgewater, NJ: Salix Pharmaceuticals Inc.; 2019.
5. Boulete IM, Thadi A, Beaufrand C, et al. *World J Gastroenterol*. 2018;24(17):1888-1900.
6. DeMicco M, Barrow L, Hickey B, et al. *Therap Adv Gastroenterol*. 2017;10(11):837-851.
7. Miner PB Jr, Koltun WD, Wiener GJ, et al. *Am J Gastroenterol*. 2017;112(4):613-621.
8. Brenner DM, Fogel R, Dorn SD, et al. *Am J Gastroenterol*. 2018;113(5):735-745.

Disclosures: D. Brenner is a consultant and speaker for Allergan/Ironwood Pharmaceuticals, for Salix Pharmaceuticals Inc, and a consultant for Shire Pharmaceuticals; H. Franklin is an employee and stockholder at Salix Pharmaceuticals Inc; G. Sayuk is a consultant and speaker for Salix Pharmaceuticals Inc, and for Allergan/Ironwood Pharmaceuticals, and a consultant for the GI Health Foundation.

Acknowledgments: Funding for this study was provided by Synergy Pharmaceuticals Inc. Poster support was funded by Salix Pharmaceuticals Inc. Medical writing and editorial support was provided by The Medicine Group (New Hope, PA, USA), in accordance with Good Publication Practice guidelines.

This poster was funded by Salix Pharmaceuticals.